Advancing Flow Cytometry for T-cell Activation and Exhaustion Analysis with Single Biomarker Cell Mimic Controls

Sarah Kotanchiyev, Kanwal Palla, Brian Kerfs, Swetha Gunturu, Kristine Valenteros

Slingshot Biosciences, Inc

SLINGSHOT BIOSCIENCES

ABSTRACT

The precise measurement of T-cell activation and exhaustion markers is crucial in immuno-oncology, vaccine development, and immune profiling. Accurate flow cytometry analysis of markers such as CD69, CD25 (activation), and PD-1, TIM-3 (exhaustion) is frequently compromised by the lack of available controls that effectively mimic these biological states, as well as variability introduced by differences in sample preparation. To address this challenge, Slingshot Biosciences developed single biomarker cell mimics expressing CD25, CD69, CD278 (ICOS), and CD38, for activation and PD-1, CTLA-4, and TIM-3 for exhaustion. We demonstrate that these cell mimics can be spiked into non-activated PBMCs and carried through the staining protocol resulting in positivity for activation and exhaustion markers. We also show similar percent positive readouts for our single biomarker controls when compared to activated PBMCs. These novel cell mimics empower researchers to monitor T-cell activation and exhaustion with greater accuracy, supporting applications ranging from immunotherapy assessments to vaccine trials.

INTRODUCTION

Slingshot leverages the principles of biochemistry, high-precision manufacturing, and polymer chemistry to engineer cell mimics that match the features of biological cells, including optical, fluorescence and biochemical properties. By addressing the limitations of biological controls, cell mimics offer a scalable, reproducible solution to accelerate and improve the development of next-generation immunotherapies.

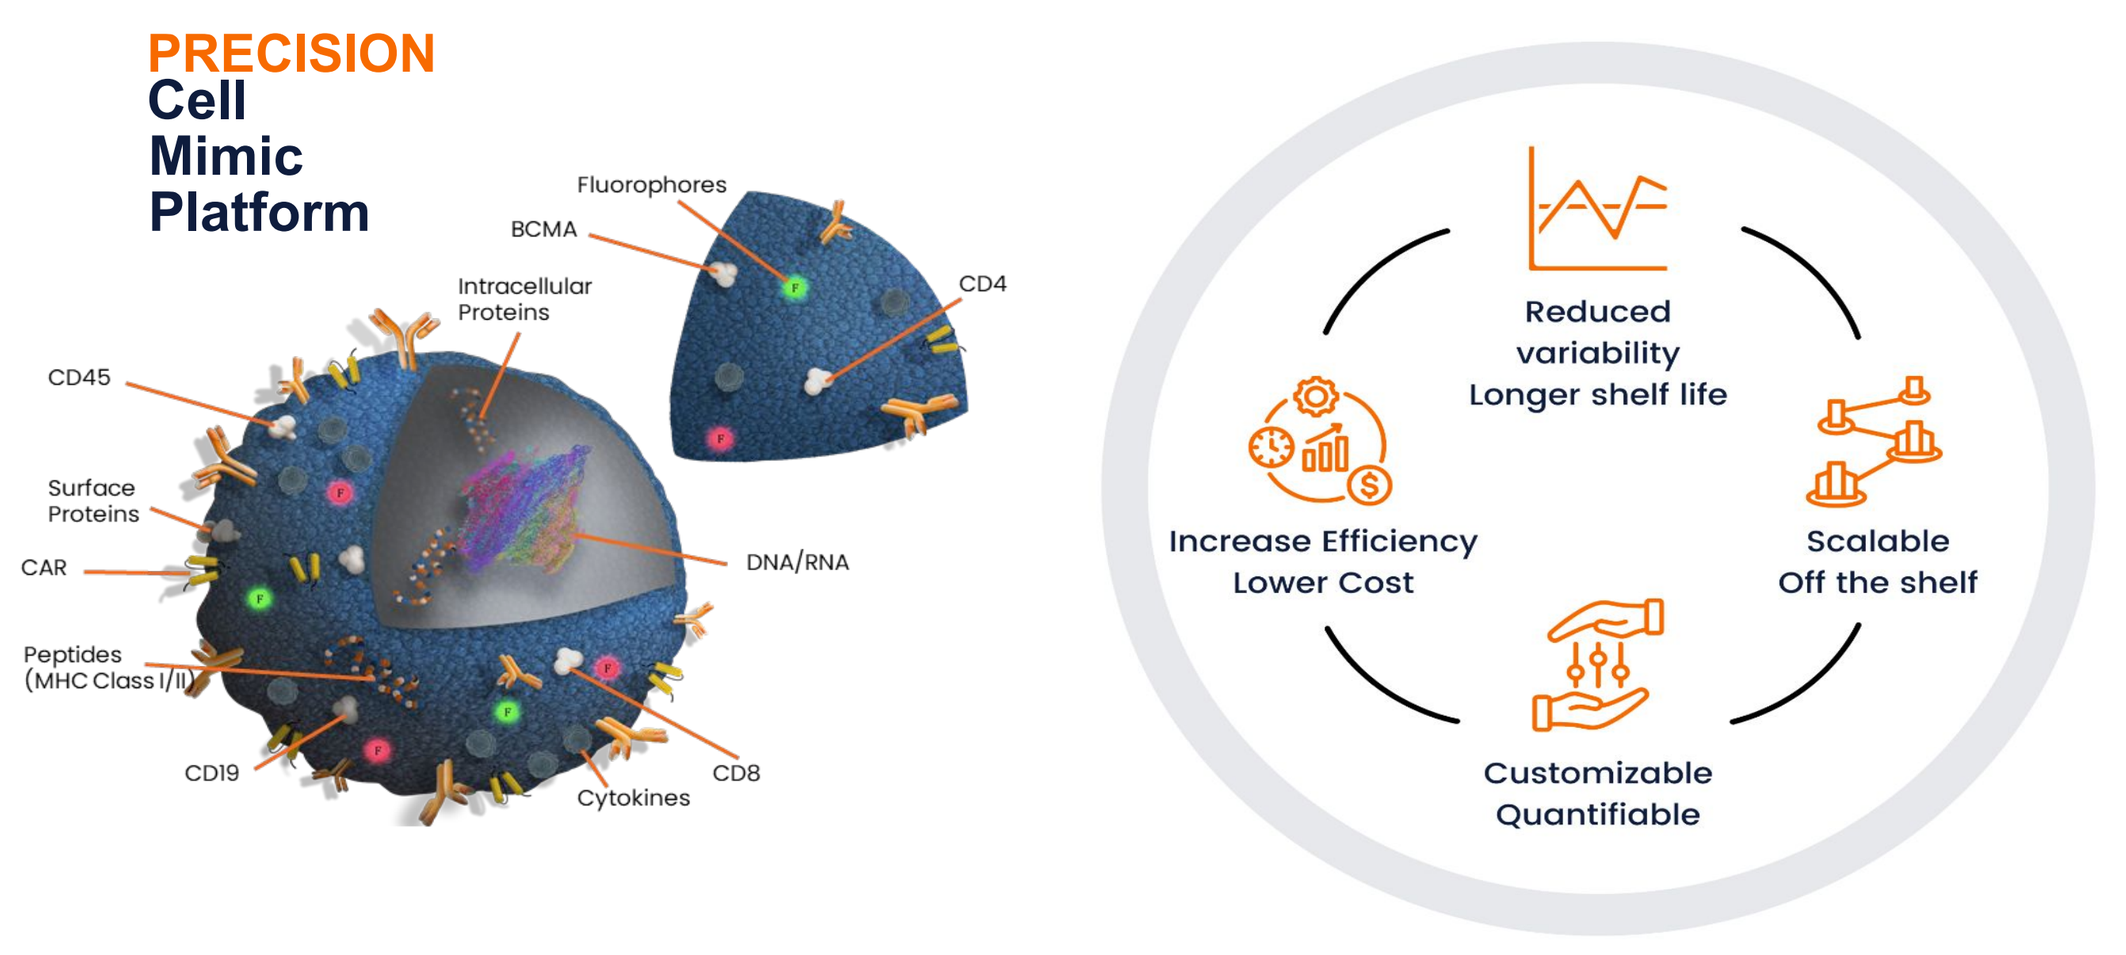
PRECISION
Cell
Mimic
Platform
Fluorophores
BCMA
Intracellular Proteins
CD4
CD45
Surface Proteins
CAR
DNA/RNA
Peptides (MHC Class I/II)
CD19
Cytokines
CD8
Reduced variability
Longer shelf life
Increase Efficiency
Lower Cost
Scalable
Off the shelf
Customizable
Quantifiable

Figure 1. Schematic of Slingshot technology capability to match biological cells and benefits for standardized assay development

Single Biomarker Activation And Exhaustion Cell Mimic Controls

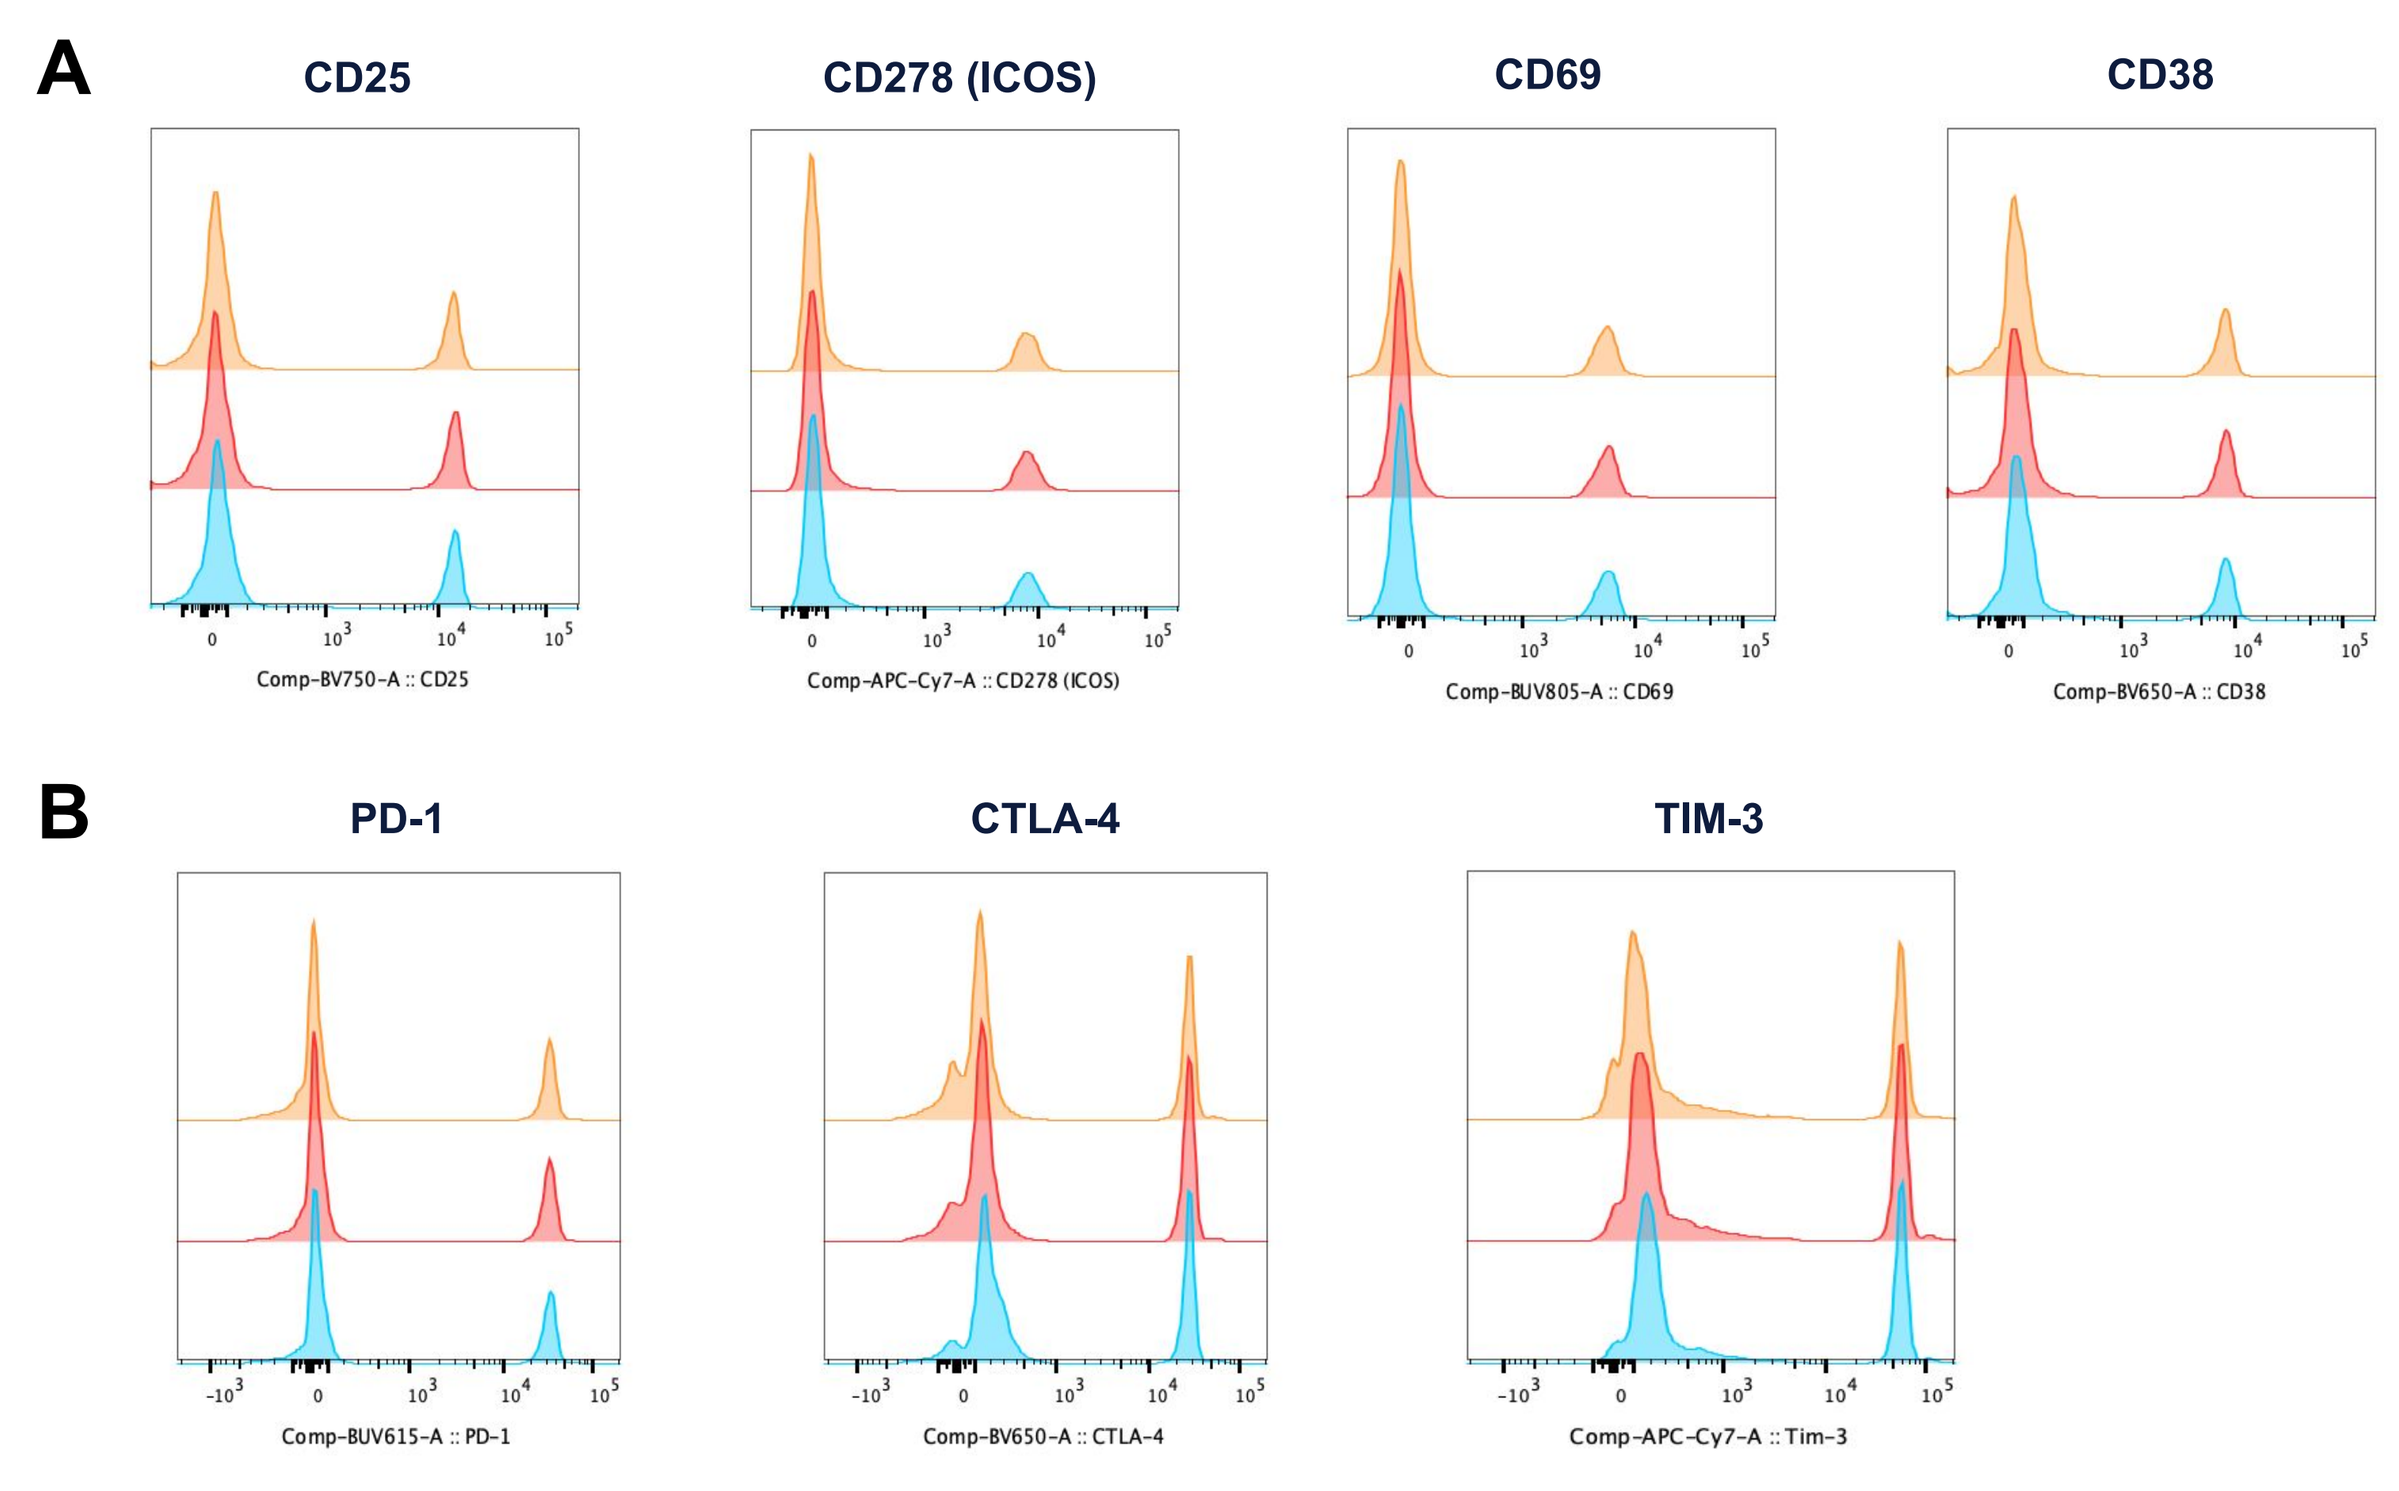
A
CD25
CD278 (ICOS)
CD69
CD38
Comp-BV750-A :: CD25
Comp-APC-Cy7-A :: CD278 (ICOS)
Comp-BUV805-A :: CD69
Comp-BV650-A :: CD38
B
PD-1
CTLA-4
TIM-3
Comp-BUV615-A :: PD-1
Comp-BV650-A :: CTLA-4
Comp-APC-Cy7-A :: Tim-3

Fig 2. Single Biomarker Cell Mimics Demonstrate Positive Expression of Activation And Exhaustion Markers. Histograms of Single Biomarker Cell Mimics spiked into PBMCs, gating only on cell mimic population to demonstrate positive expression of **A.** Activation and **B**. Exhaustion Markers. Histogram data represents three replicates for each marker.

Single Biomarker Cell Mimic Controls Can Be Spiked Into Peripheral Blood Mononuclear Cells

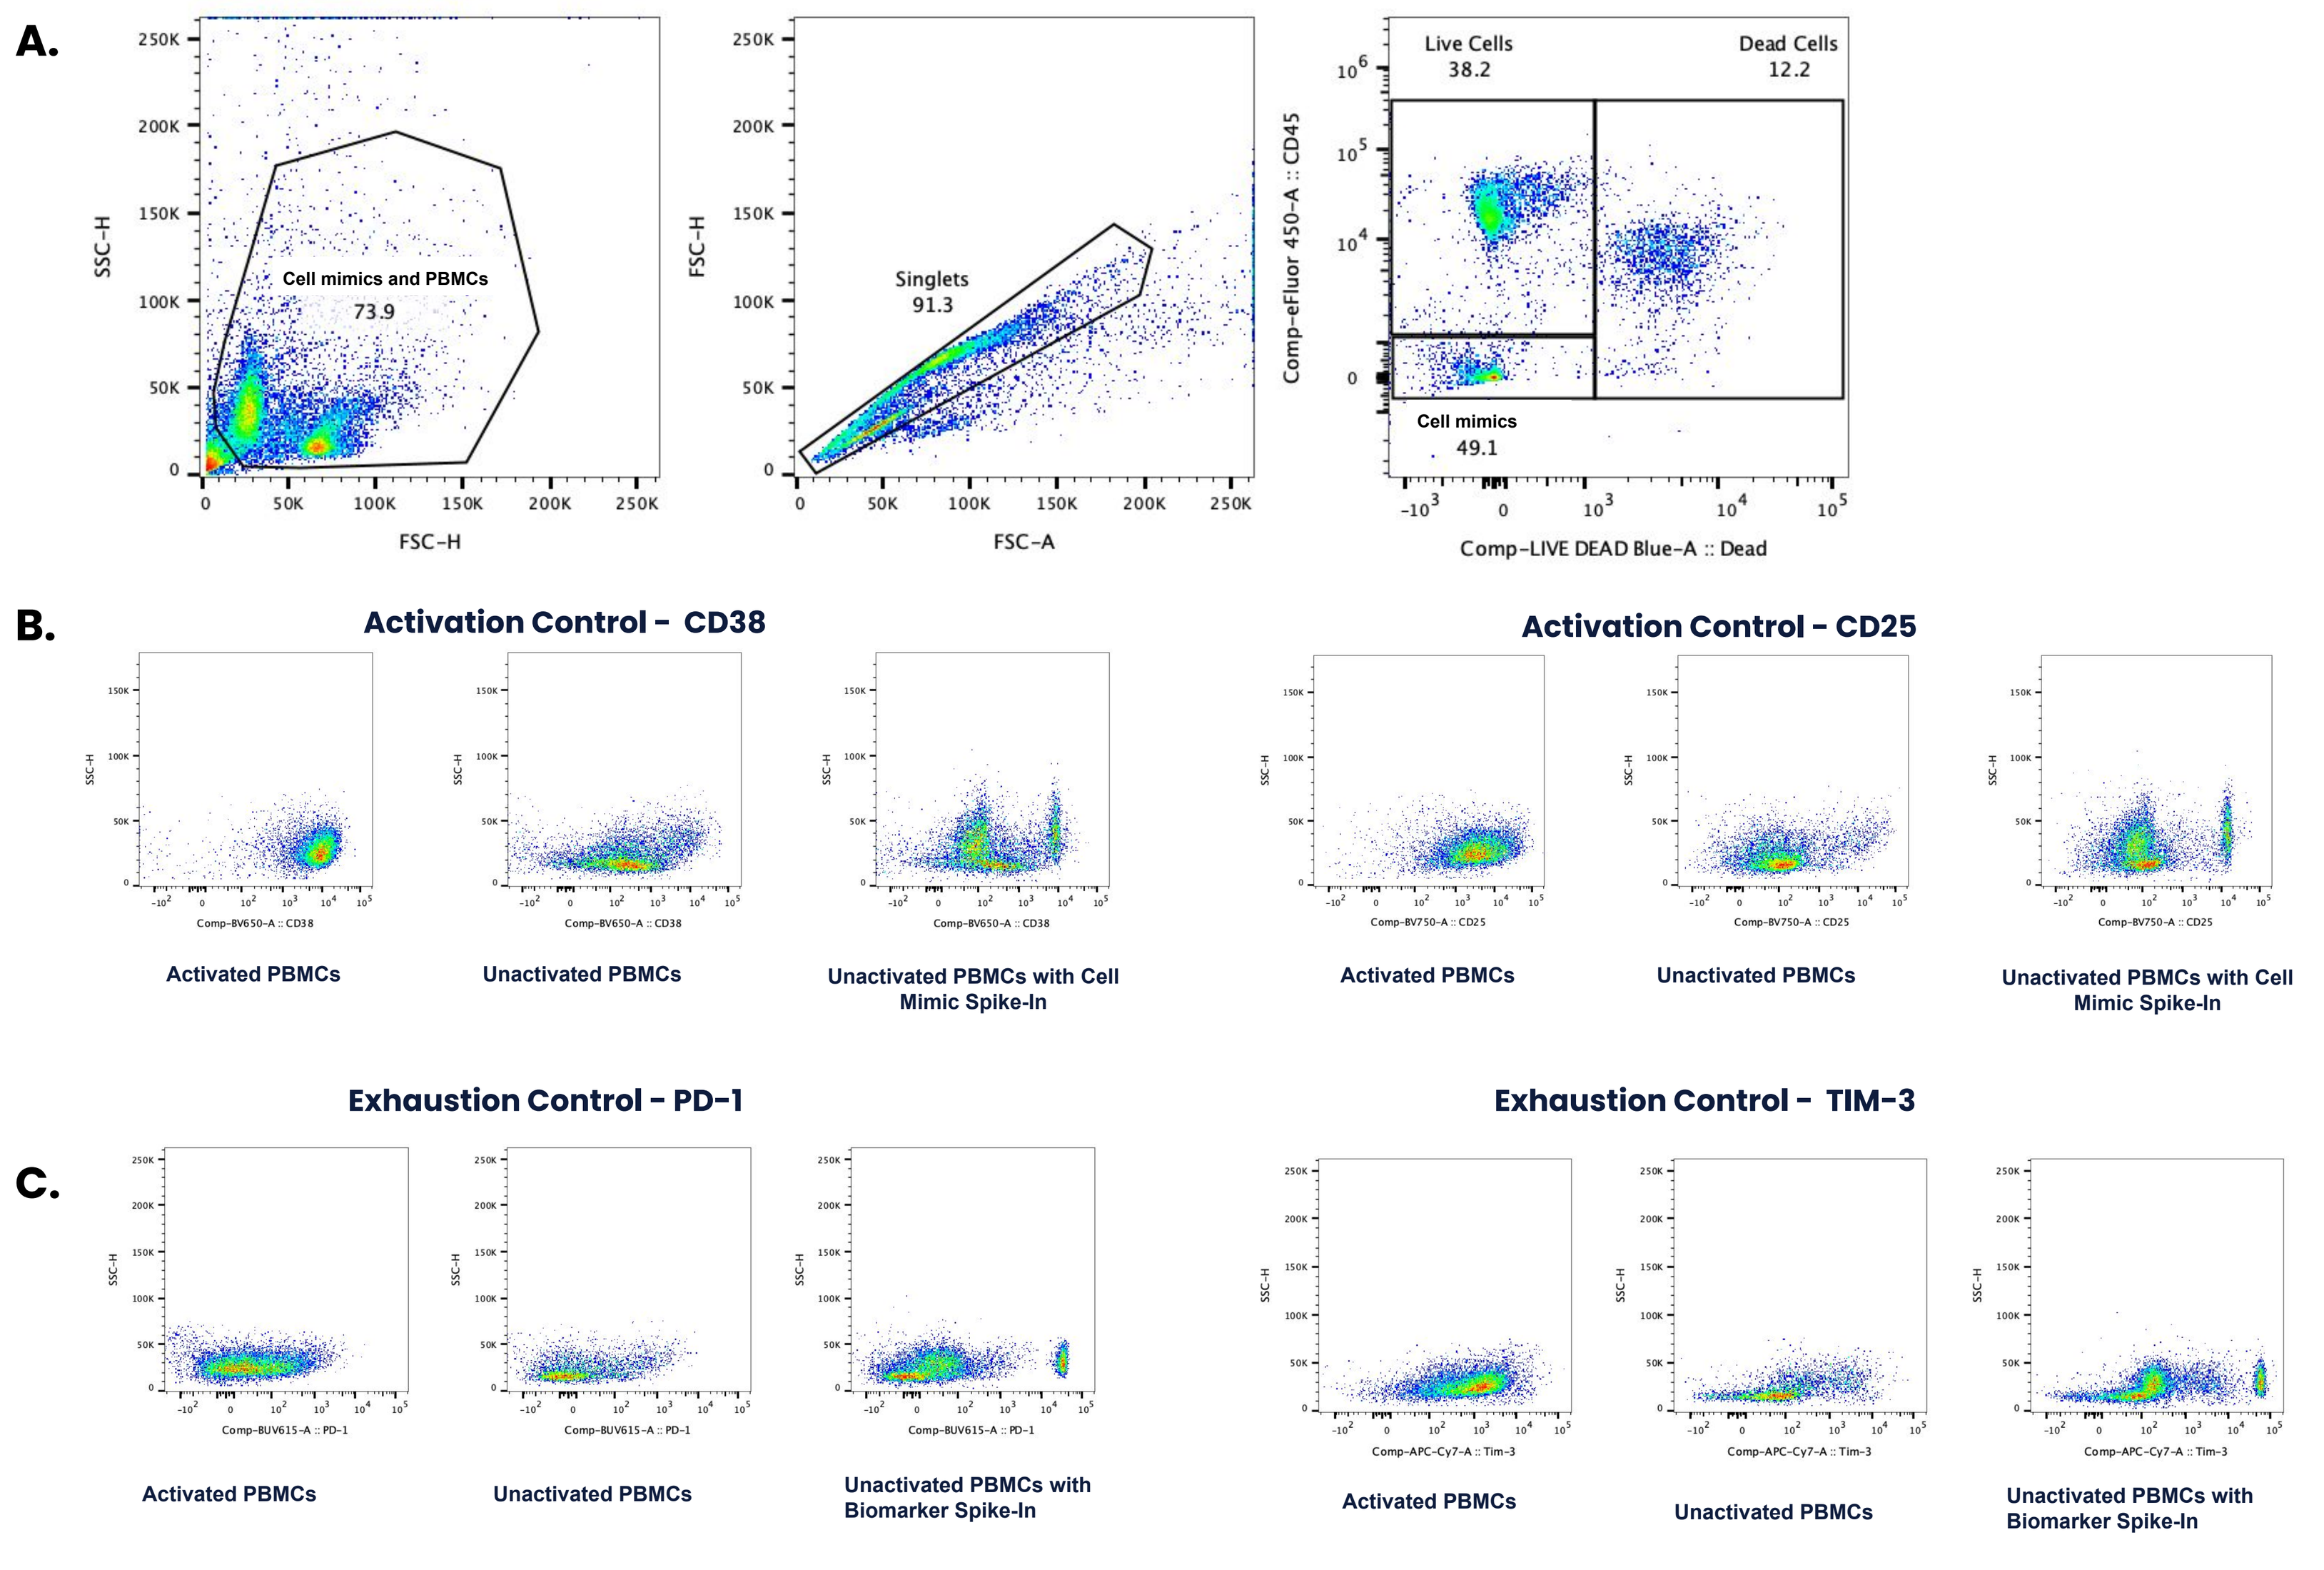
A.
Cell mimics and PBMCs
73.9
SSC-H
FSC-H
Singlets
91.3
FSC-A
Live Cells
38.2
Dead Cells
12.2
Cell mimics
49.1
Comp-eFluor 450-A :: CD45
Comp-LIVE DEAD Blue-A :: Dead
B.
Activation Control - CD38
Activation Control - CD25
Activated PBMCs
Unactivated PBMCs
Unactivated PBMCs with Cell Mimic Spike-In
C.
Exhaustion Control - PD-1
Exhaustion Control - TIM-3
Activated PBMCs
Unactivated PBMCs
Unactivated PBMCs with Biomarker Spike-In

Fig 3. Single Biomarker Cell Mimic Controls Can Be Spiked Into PBMCs to Represent Exhausted or Activated States

A. Gating scheme showing how cell mimics are represented based on optical parameters on a flow cytometer and gated out from live vs. dead cells.

B. Unactivated PBMCs were spiked with single biomarker cell mimics expressing CD38 or CD25 to represent activated cells.

C. Unactivated PBMCs were spiked with single biomarker cell mimics expressing PD-1 or TIM-3 to represent exhausted cells.

Development Of Multi-Biomarker Cell Mimics Demonstrating T, B and NK Cell Populations

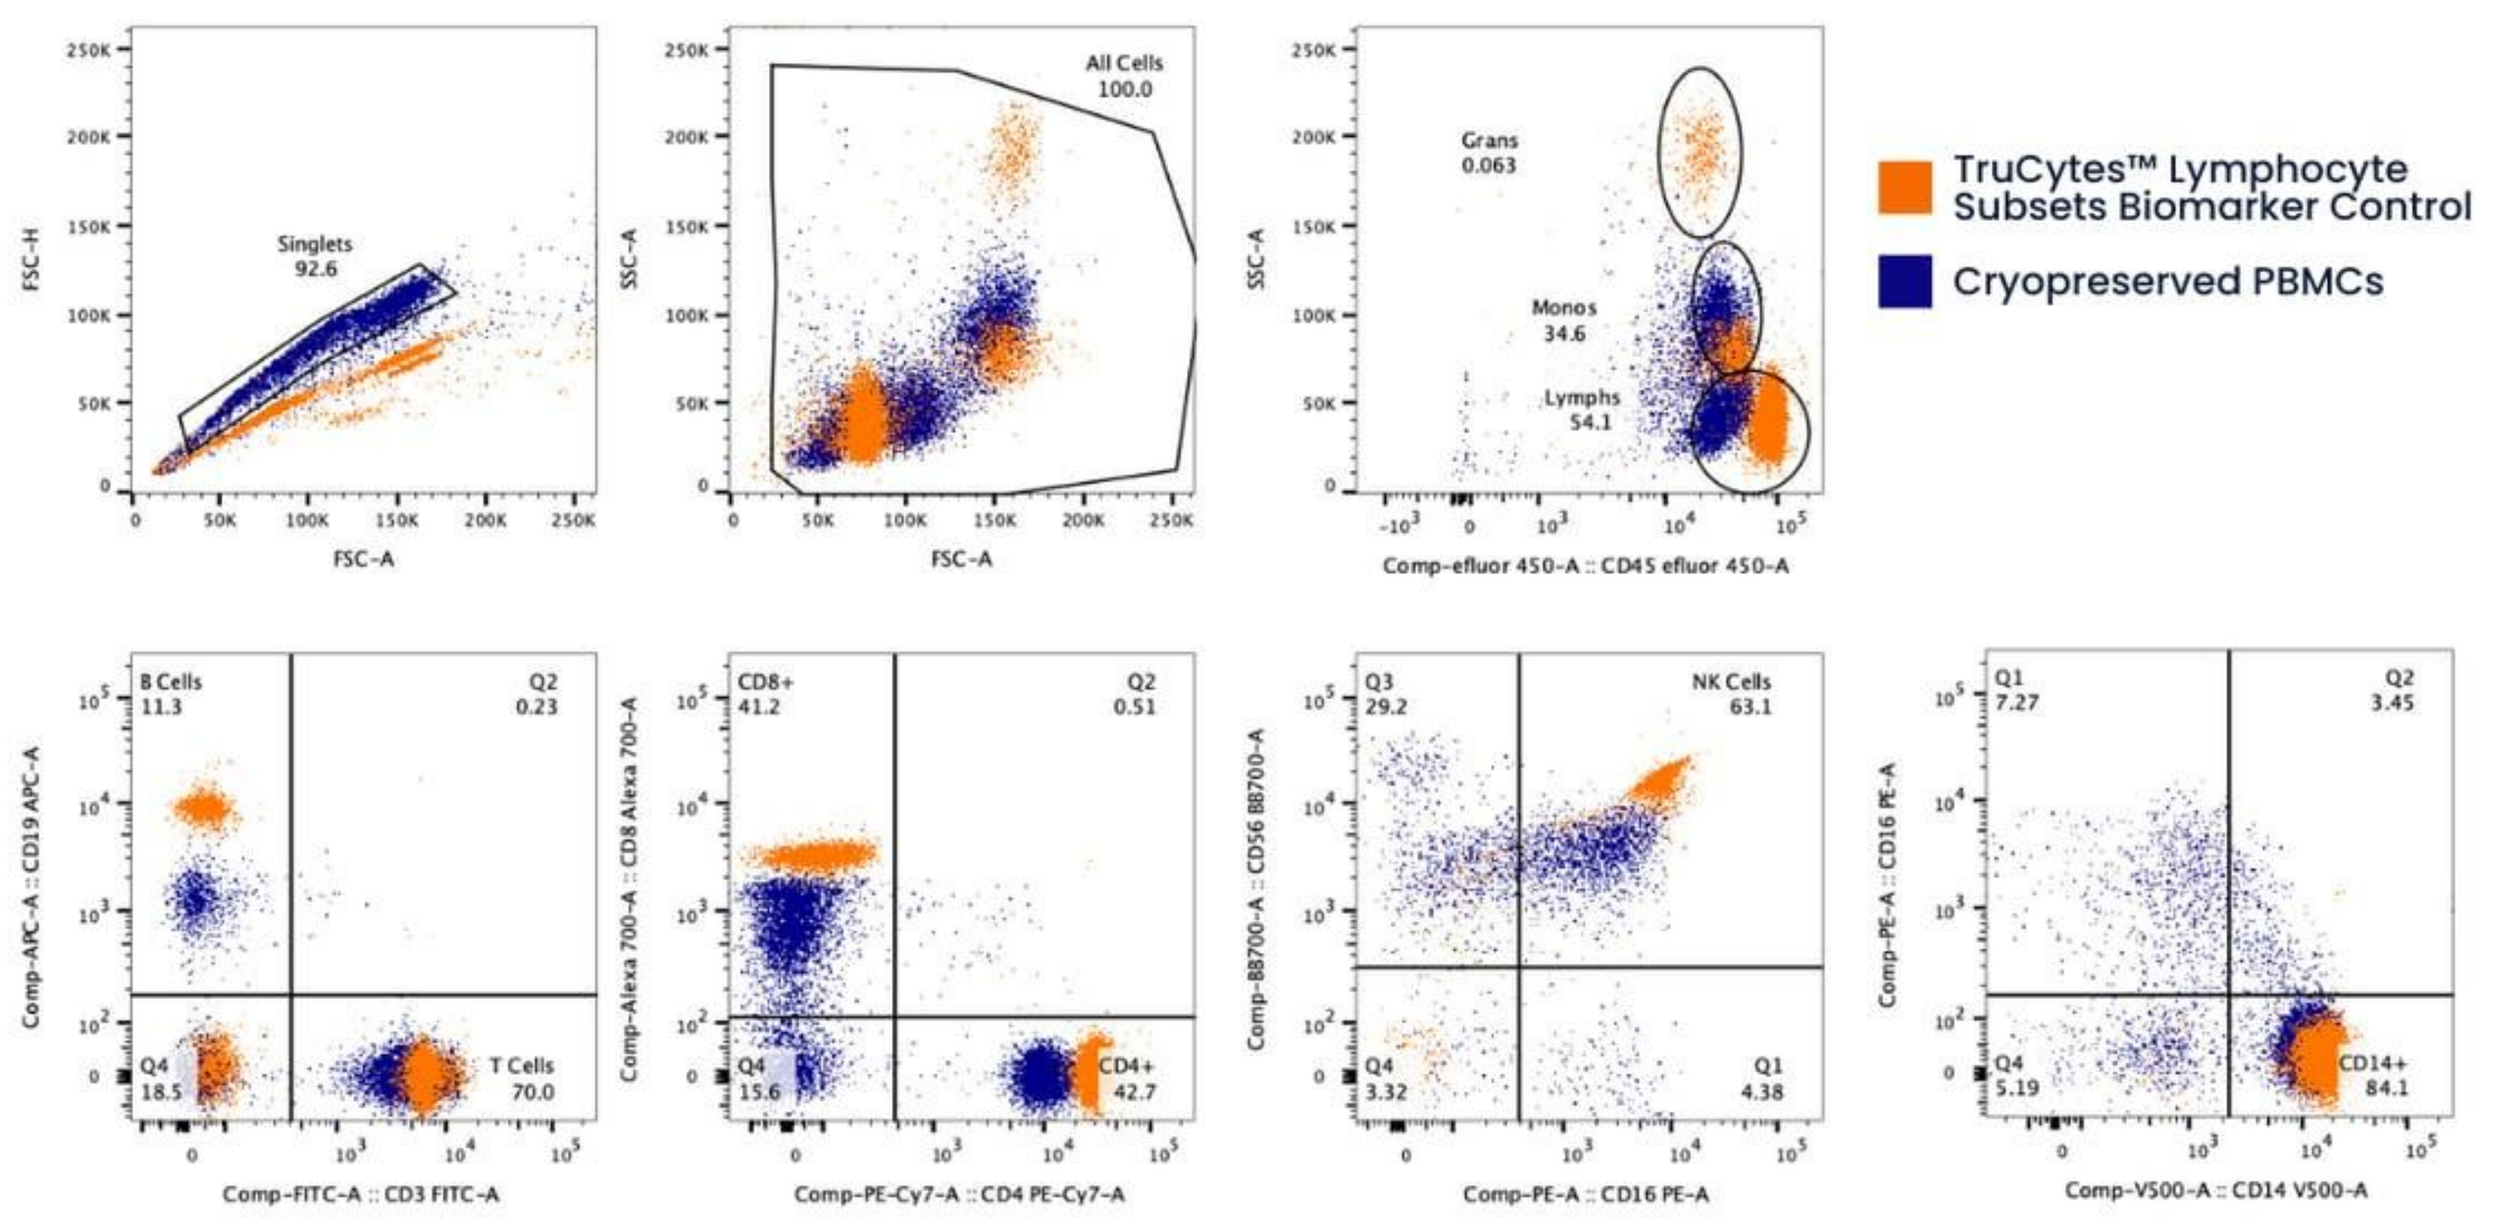
Singlets
92.6
All Cells
100.0
Grans
0.063
Monos
34.6
Lymphs
54.1
TruCytes™ Lymphocyte Subsets Biomarker Control
Cryopreserved PBMCs
B Cells
11.3
T Cells
70.0
CD8+
41.2
CD4+
42.7
NK Cells
63.1
CD14+
84.1

Figure 4. Slingshot TruCytes™ Lymphocytes Subsets Control demonstrates similar optical properties to cryopreserved peripheral blood mononuclear cells (PBMCs)

Pre Approved Biomarkers for Custom Solutions*

T Cell Markers	CD2, CD3, CD4, CD5, CD7, CD8, CD25, CD27, CD28, CD45, CD134 (OX40), CD137, CD152 (CTLA-4), CD154 (CD40L), CD278 (ICOS), CD279 (PD-1)
B Cell Markers	CD19, CD20, CD22, CD23, CD24, CD38, CD79b, CD86, CD267 (TACI), CD268 (BAFF-R), CD269 (BCMA)
Myeloid Markers	CD14, CD16, CD33, CD64, CD66, CD68, CD163, CD172a (SIRPα)
Stem/Progenitor Markers	CD34, CD117 (c-Kit), CD123 (IL-3Rα), CD135 (FLT3)
NK Cell Markers	CD16, CD56, CD335 (NKp46)
Activation Markers	CD25, CD69, CD71 (Transferrin Receptor), CD134 (OX40), CD137, CD278 (ICOS), CD80, CD86
Adhesion Molecules	CD54 (ICAM-1), CD58 (LFA-3), CD102 (ICAM-2), CD166 (ALCAM)
Immune Checkpoint Markers	CD152 (CTLA-4), CD279 (PD-1), CD274 (PD-L1), CD223 (LAG-3), TIM-3
Cytokine Receptors	CD120b (TNF-RII), CD122 (IL-2Rβ), CD124 (IL-4R), CD126 (IL-6R), CD127 (IL-7Rα), CD213a1 (IL-13Rα1), CD213a2 (IL-13Rα2), CD215 (IL-15Rα), CD217 (IL-17R), CD218a (IL-18Rα), CD218b (IL-18Rβ)
Tumor/Proliferation Markers	Ki-67 (MKI67), CD71 (Transferrin Receptor), CD99, CD227 (MUC1), CD340 (HER2/neu), LRRC15
Other Functional Markers	EGFR, CD36, CD39, CD40, CD47, CD52, CD59, CD93, CD109, CD112, CD147, CD150, CD155, CD162, CD203c, CD221, CD229, CD252 (OX40L), CD254 (RANKL), CD257 (APRIL), CD304 (Neuropilin-1), CD314 (NKG2D), CD319 (SLAMF7), CD328 (Siglec-7), CD329 (Siglec-9)
Transcription Factors	FOXP3, ROR1
Cytokines	IL-2, IL-4, IL-21, TNFa, TGFbR2

*Other markers can be evaluated

Table 1. Slingshot evaluated biomarkers for custom projects. These markers can be added to Slingshot cell mimics to develop customizable controls to meet unique assay needs.

CONCLUSIONS

Slingshot Biosciences has developed precision-engineered cell mimics expressing key activation and exhaustion biomarkers as well as key immune cell markers, as scalable, reproducible quality controls. These cell mimics offer several key advantages:

- Specific biomarker expression detectable by flow cytometry with bright signal intensity
- When single biomarker controls are spiked into PBMC samples, they closely replicate the optical and phenotypic profiles of activated or exhausted T cells
- Match PBMCs in size and internal complexity, ensuring compatibility with established gating schemes in immune cell analysis workflows
- Seamlessly integrate into standard antibody staining protocols