Cell Mimics as Reliable Controls for Consistent, Reproducible Flow Cytometry Assays in Cell Therapy

Sarah Kotanchiyev, Swetha Gunturu, Subhanip Biswas, Louisa D'Lima, Armando Martinez, Kanwal Palla
Slingshot Biosciences Inc, Emeryville, CA, 94608, USA.

ABSTRACT

Background:
Reliable and consistent quality control remains a major challenge in cell therapy development, particularly in validating flow cytometry assays where traditional biological controls such as PBMCs or cell lines are often variable. Here, we demonstrate a solution using polymer-based cell mimics to represent T, B, and NK cell subsets from healthy donor blood, designed to minimize variability in assay performance.

Methods:
Two lots of lyophilized and cryopreserved commercially-available PBMC controls were evaluated for lot-to-lot consistency against TruCytes™ Lymphocyte Subset Cell Mimic Controls using a Cytek® Aurora CS. Fixation buffers from three vendors were tested on TruCytes™ for antigen-specific antibody binding post-fixation. Additionally, TruCytes™cell mimics were tested with two RBC lysis protocols to assess compatibility with a 1X RBC Lysis Buffer. Linearity studies were performed by serial dilution of TruCytes Lymphocytes Subsets Control and staining with an in-house antibody panel to identify expression of key populations by flow cytometry.

Results:
Our data show that these cell mimics offer consistent lot-to-lot reproducibility and stability far exceeding that of lyophilized and cryopreserved PBMC controls. We measured the percentage of key cell types and expression of cellular markers across lots and determined that cell mimics maintained accuracy, with CVs of 0.1% to 5.7%, significantly outperforming PBMC-based controls that showed CVs ranging from 1.6% to 36.6%. Cell mimics were also shown to be compatible with existing workflows using common lyse and fixative buffers. We also demonstrated that our cell mimics were compatible with commercially available antibody panels. Linearity assessments demonstrated strong correlation between measured and expected values with R^2 values of 0.995 or 0.996 for monocytes, lymphocytes and granulocytes populations.

Conclusion:
Leveraging cell mimics can help standardize and accelerate patient and donor characterization and qualification, providing a robust quality control solution that meets the high standards required for cell therapy development.

TruCytes™ Lymphocytes Subsets Control Cell Mimics as Reliable Alternative to Donor-Derived Controls

Slingshot Biosciences aims to solve variability concerns with donor derived controls by providing the first-of-its-kind TBNK cell mimics. Our TruCytes™ Lymphocyte Subset control is a lyophilized mixture of cell mimics engineered to mimic PBMCs or leukopak-based controls in terms of scatter, biomarker expression levels, and immune cell subset population frequencies.

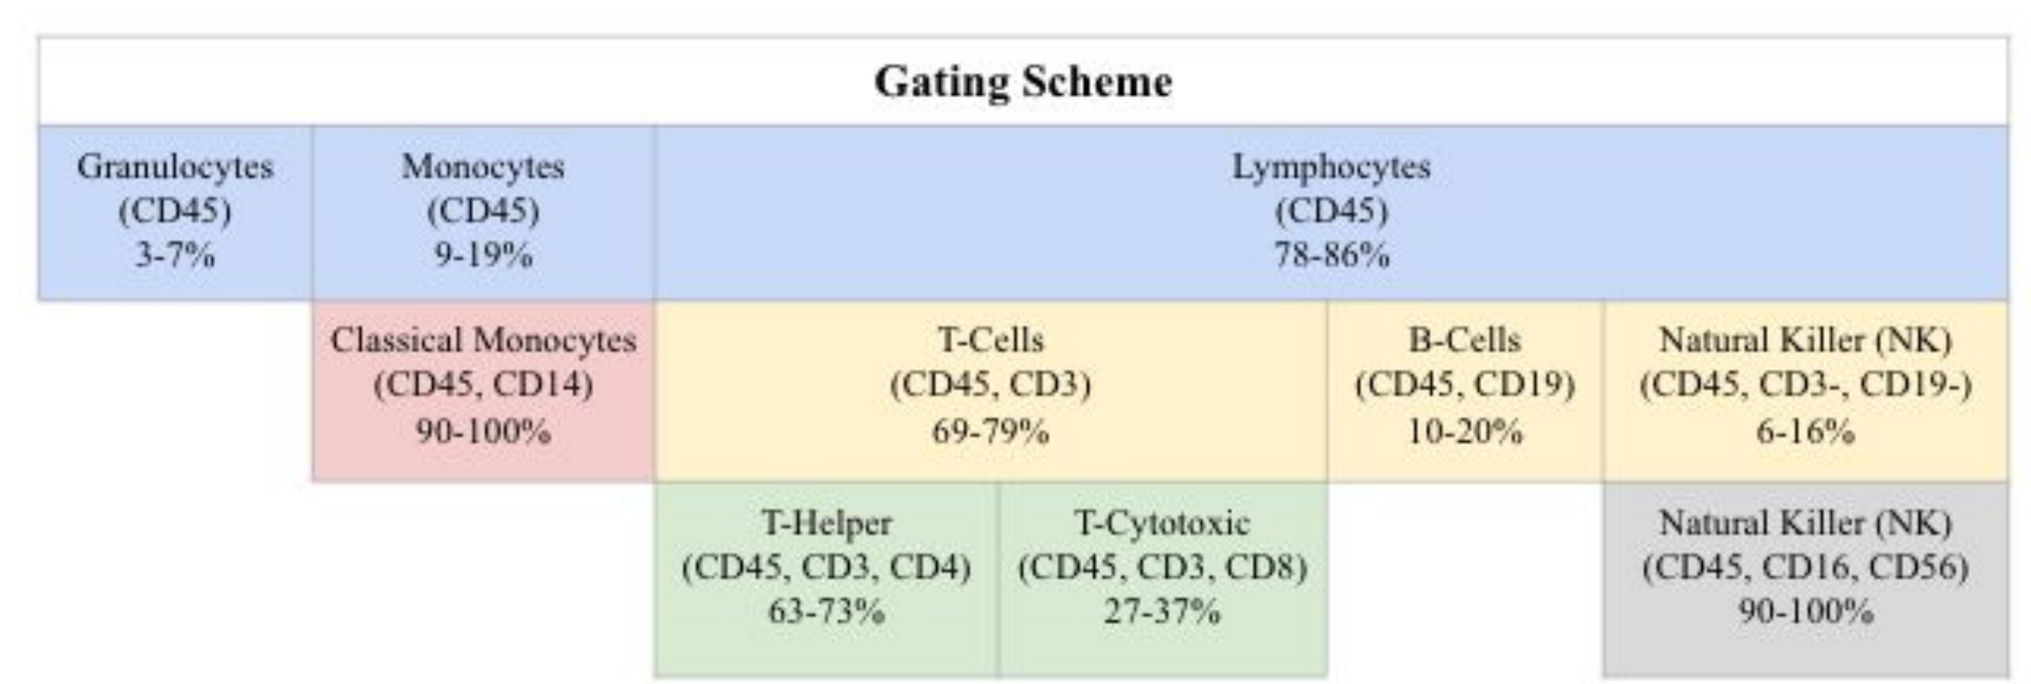
Gating Scheme
Granulocytes (CD45) 3-7%
Monocytes (CD45) 9-19%
Lymphocytes (CD45) 78-86%
Classical Monocytes (CD45, CD14) 90-100%
T-Cells (CD45, CD3) 69-79%
B-Cells (CD45, CD19) 10-20%
Natural Killer (NK) (CD45, CD3-, CD19-) 6-16%
T-Helper (CD45, CD3, CD4) 63-73%
T-Cytotoxic (CD45, CD3, CD8) 27-37%
Natural Killer (NK) (CD45, CD16, CD56) 90-100%

Figure 1. The populations included in TruCytes™ Lymphocyte Subset are T cells (CD3 and CD4/CD8), NK cells (CD16 and CD56), B cells (CD19), and Monocytes (CD14).

Lot-to-Lot Reproducibility Assessment

%CV across two lots of each control type				
Population	Lyophilized PBMCs Lots	Cryopreserved PBMCs Lots	Lyophilized Lymphocytes Lots	TruCytes™ Lymphocytes Subsets Lots
Grans CD45+	NA	NA	N/A	2.5
Monos CD45+	13.9	34.5	N/A	4.4
Lymphs CD45+	10.2	15.2	0.9	0.6
T Cells CD3+	12.1	1.2	0.5	1.3
B Cells CD19+	36.6	19.8	6.5	5.7
NK Cells CD3-CD19-	29.1	8.8	7.7	1.6
T Helper CD4+	15.2	17.4	0.8	1.2
T Cytotoxic CD8+	18.7	18.7	4.3	2.3
NK Cells CD16+ CD56+	17.7	3.3	3.9	0.3
Classical Monos CD14+	1.6	8.3	N/A	0.1

Table 1. TruCytes™ Lymphocyte Subsets demonstrate less variation in % populations between lots than PBMCs. % CV was calculated for the percent population as evaluated on the Cytek Aurora across two lots of each different control material. Red indicates high variability. Blue indicates low variability.

Linearity assessment of TruCytes Lymphocytes Subsets Populations

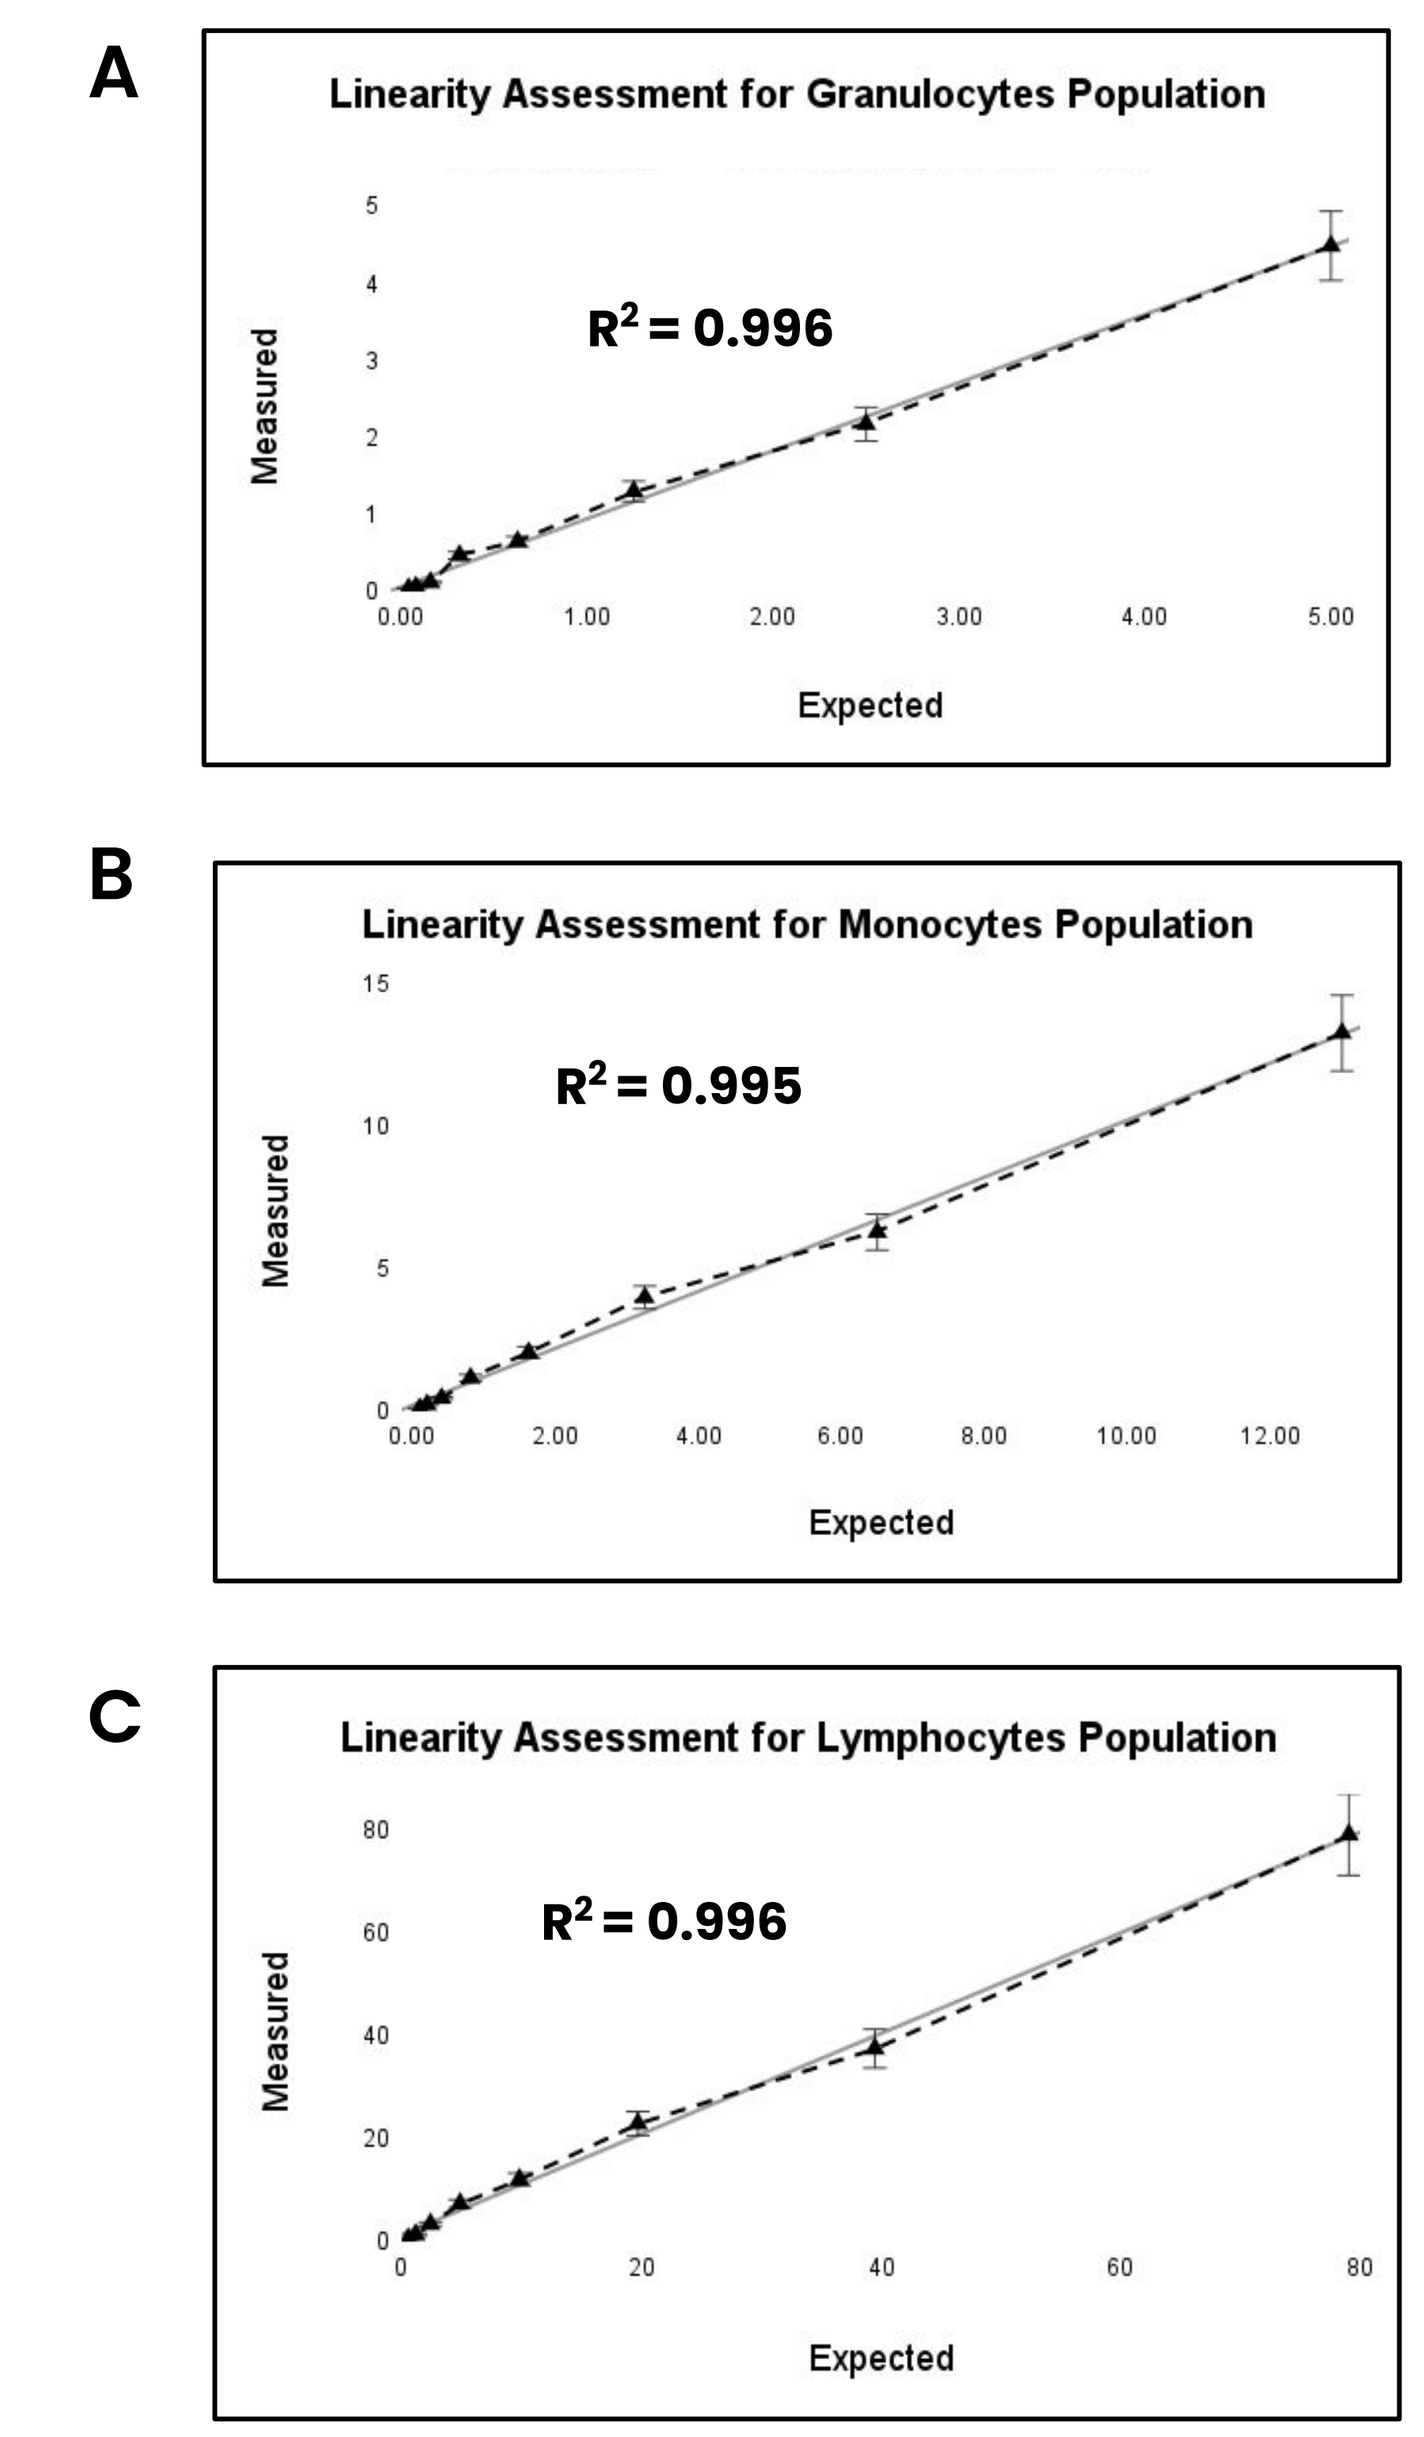
A
Linearity Assessment for Granulocytes Population
$R^2 = 0.996$
Measured
Expected
B
Linearity Assessment for Monocytes Population
$R^2 = 0.995$
Measured
Expected
C
Linearity Assessment for Lymphocytes Population
$R^2 = 0.996$
Measured
Expected

Figure 3. Linearity of TruCytes Lymphocyte Subset Control
Serial dilution linearity assessment of key immune populations **A**. Granulocytes **B**. Monocytes and **C**. Lymphocytes demonstrate robust linearity

TruCytes™ Lymphocyte Subset Control are Compatible with Common Workflows and Buffers

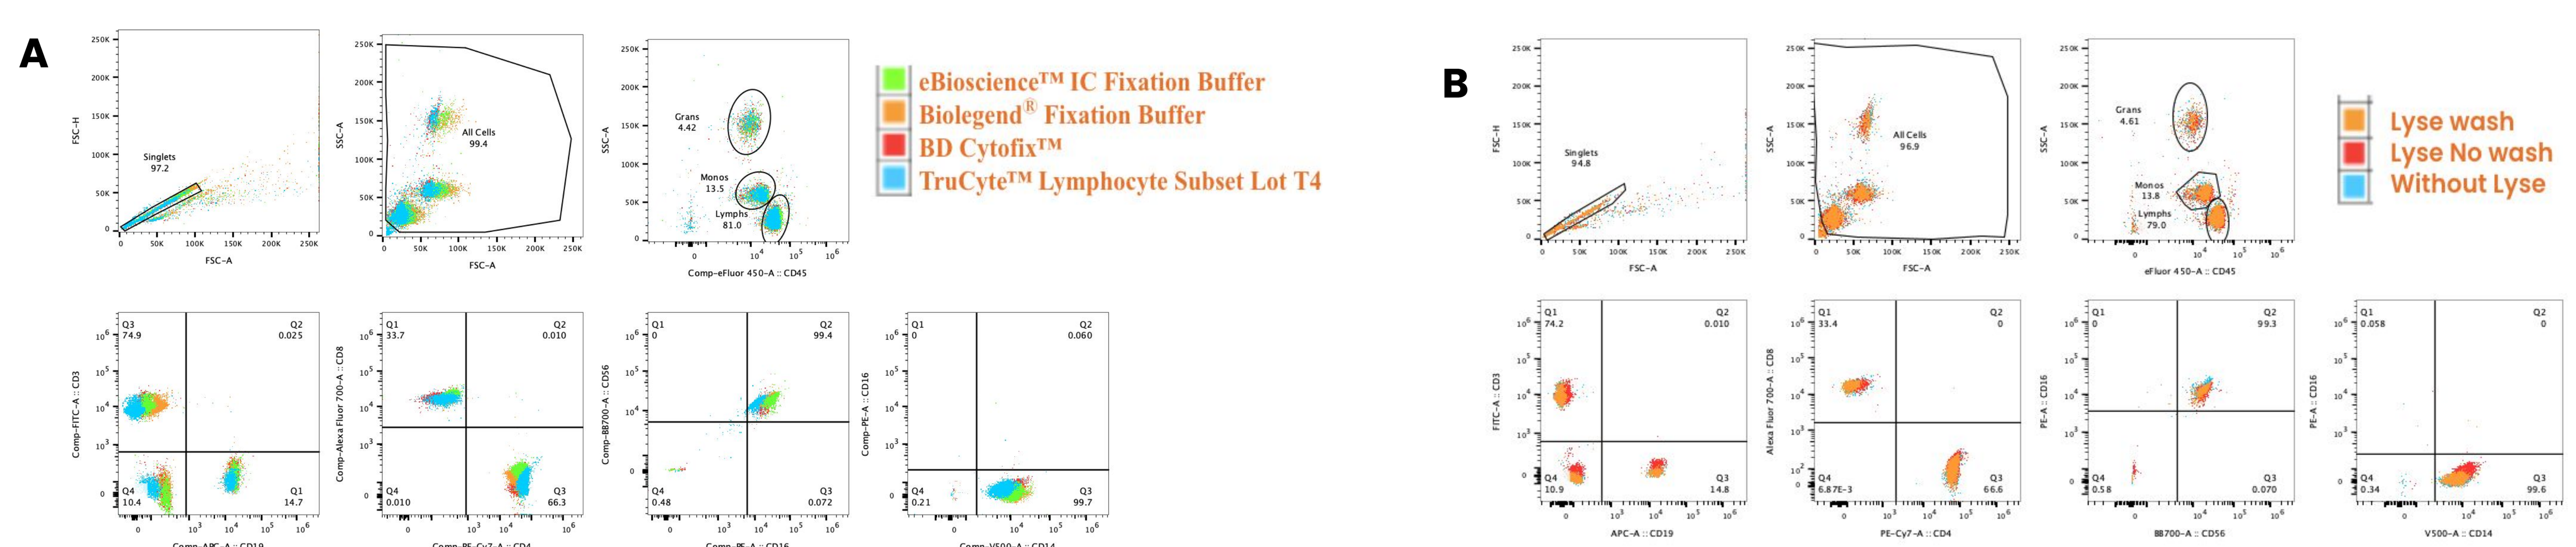
A
eBioscience™ IC Fixation Buffer
Biolegend® Fixation Buffer
BD Cytofix™
TruCyte™ Lymphocyte Subset Lot T4
B
Lyse wash
Lyse No wash
Without Lyse

Figure 2. TruCytes™ Lymphocyte Subset control demonstrate compatibility with **A**. common commercially available fixation buffers and **B**. lyse/wash or lyse/no wash protocols.

Population	Slingshot's TruCytes Lymphocytes Subsets ranges (%)	Data from Slingshot's In-house Panel (%)	Beckman Coulter DURAClone IM Kit (%)	BD Multitest 6-color Kit (%)	BioLegend Human TBNK 4-color Kit (%)	%CV between different kits
Grans CD45+	3-7	5	4	5	4	12.8
Monos CD45+	9-19	13	12	13	13	3.9
Lymphs CD45+	78-86	79	81	81	81	1.2
T Cells CD3+	69-79	75	74	74	75	0.8
B Cells CD19+	10-20	15	15	15	15	0.0
NK Cells CD3-CD19-	6-16	11	11	11	11	0.0
T Helper CD4+	63-73	67	66	66	Marker not in panel	0.9
T Cytotoxic CD8+	27-37	33	34	34	Marker not in panel	1.7
NK Cells CD16+CD56+	90-100	99	100	95	98	2.2
Classical Monos CD14+	90-100	99	100	Marker not in panel	Marker not in panel	NA

Compatibility of TruCytes Lymphocytes Subsets Control with Commercially Available Antibody Panels
Table 2. TruCytes Lymphocyte Subset Control demonstrates compatibility with commercial antibody kits, showing low CVs across all the percent populations and antibody panels.

SUMMARY

TruCytes™ Lymphocyte Subset cell mimics can be easily adopted as a reliable, cost-effective alternative to PBMC-based controls in immunophenotyping assays as determined by:

- Consistency in lot-to-lot performance compared to PBMC-based immunophenotyping controls showing a range of 0.1% - 5.7% CVs versus 1.6% - 36.6% CVs for PBMC controls.
- Compatibility with existing workflows using common lyse and fixative buffers.
- Exhibits linear dilution at a range of dilutions.
- Compatibility with commercially available antibody kits.