Development and Characterization of Synthetic Extracellular Vesicle (EV) Mimics for Enhanced Flow Cytometry Standardization

Daixuan Zhang, Annalise Barnette
Slingshot Biosciences, Inc.

SLINGSHOT BIOSCIENCES

ABSTRACT

Flow cytometry analysis of single extracellular vesicles (EVs), particularly exosomes, is gaining traction in diagnostics and therapeutics due to their ability to carry complex cargo and to function as biomarkers. EV specific standard reference materials are vital in this process, enabling standardization, cross-study comparison and assay validation. Currently available synthetic controls, such as polystyrene and silica nanoparticles, have comparatively high refractive indices (RI = 1.63 and 1.47) to natural EVs (RI = 1.36-1.40). This mismatch in optical property results in inaccurate size designations and variability across different instruments and assays.

To address this shortcoming, Slingshot Biosciences has developed synthetic polymer-based EV mimics with EV-matching RI (~1.38), and nominal diameter ranging from 100nm to 400nm. These EV mimics are homogenous and monodisperse, as characterized by DLS, TEM, SEM and Cryo-TEM measurements. Additionally, our EV mimics can be conjugated with various fluorophores, enabling multiparametric analysis in commonly used fluorescent channels (e.g. FITC, PE and APC) and more reliable detection in flow cytometry. We demonstrate that the fluorescence signals are brighter and more consistent than off-the-shelf stained human EVs.

Preliminary validation through flow cytometry and superresolution microscopy has demonstrated the robust performance of our EV mimics for EV biomarker analysis, size determination, and quality control (QC). They represent a promising advancement in flow cytometry standardization to support the growing demands of EV-based diagnostics and therapeutics.

METHODOLOGY

- Various fluorophores are conjugated to Slingshot Biosciences' proprietary EV mimics.
- RI measurements were performed by Nanostics, Inc, on their ClarityDX® platform, calibrated with Cargille RI oil standards.
- Super-resolution microscopy was performed by ONI with their Nanoimager instrument.
- Flow cytometry assays were performed with a Beckman Coulter Cytoflex S flow cytometer. Side scatter was evaluated with 405 nm violet laser. Concentration of EV mimics was titrated with pre-filtered buffer to avoid swarm detection. Fluorescence threshold was used to minimize background noise.

EV-like Size and Refractive Index

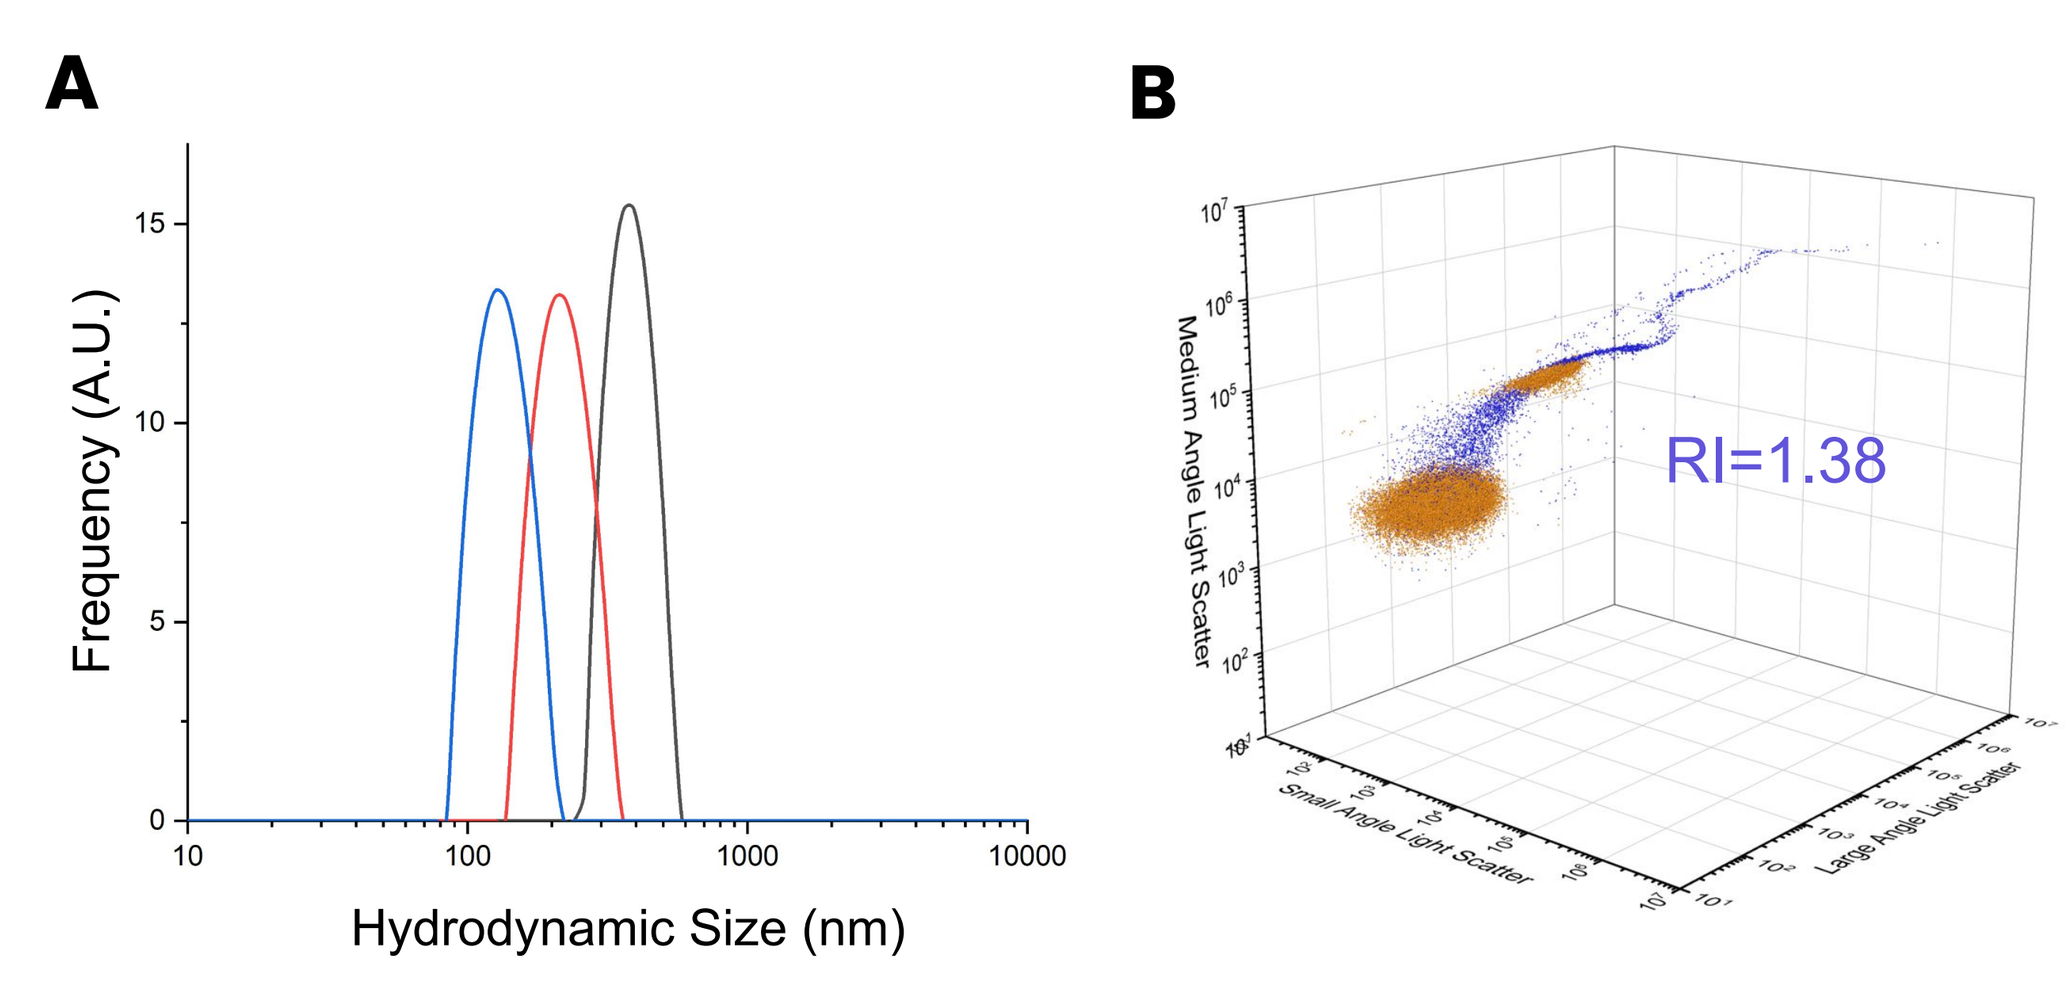
A
B
Frequency (A.U.)
Hydrodynamic Size (nm)
RI=1.38
Medium Angle Light Scatter
Small Angle Light Scatter
Large Angle Light Scatter

Figure 1: **A.** Hydrodynamic size and distribution of small, medium and large (~100 nm, 200 nm, and 400 nm) EV mimics measured by DLS. **B.** Refractive index of large synthetic EV mimics (Brown) measured using MALS, compared to oil standard emulsion with known RI = 1.38 (Blue).

True-to-life Scatter with Precise Fluorescence

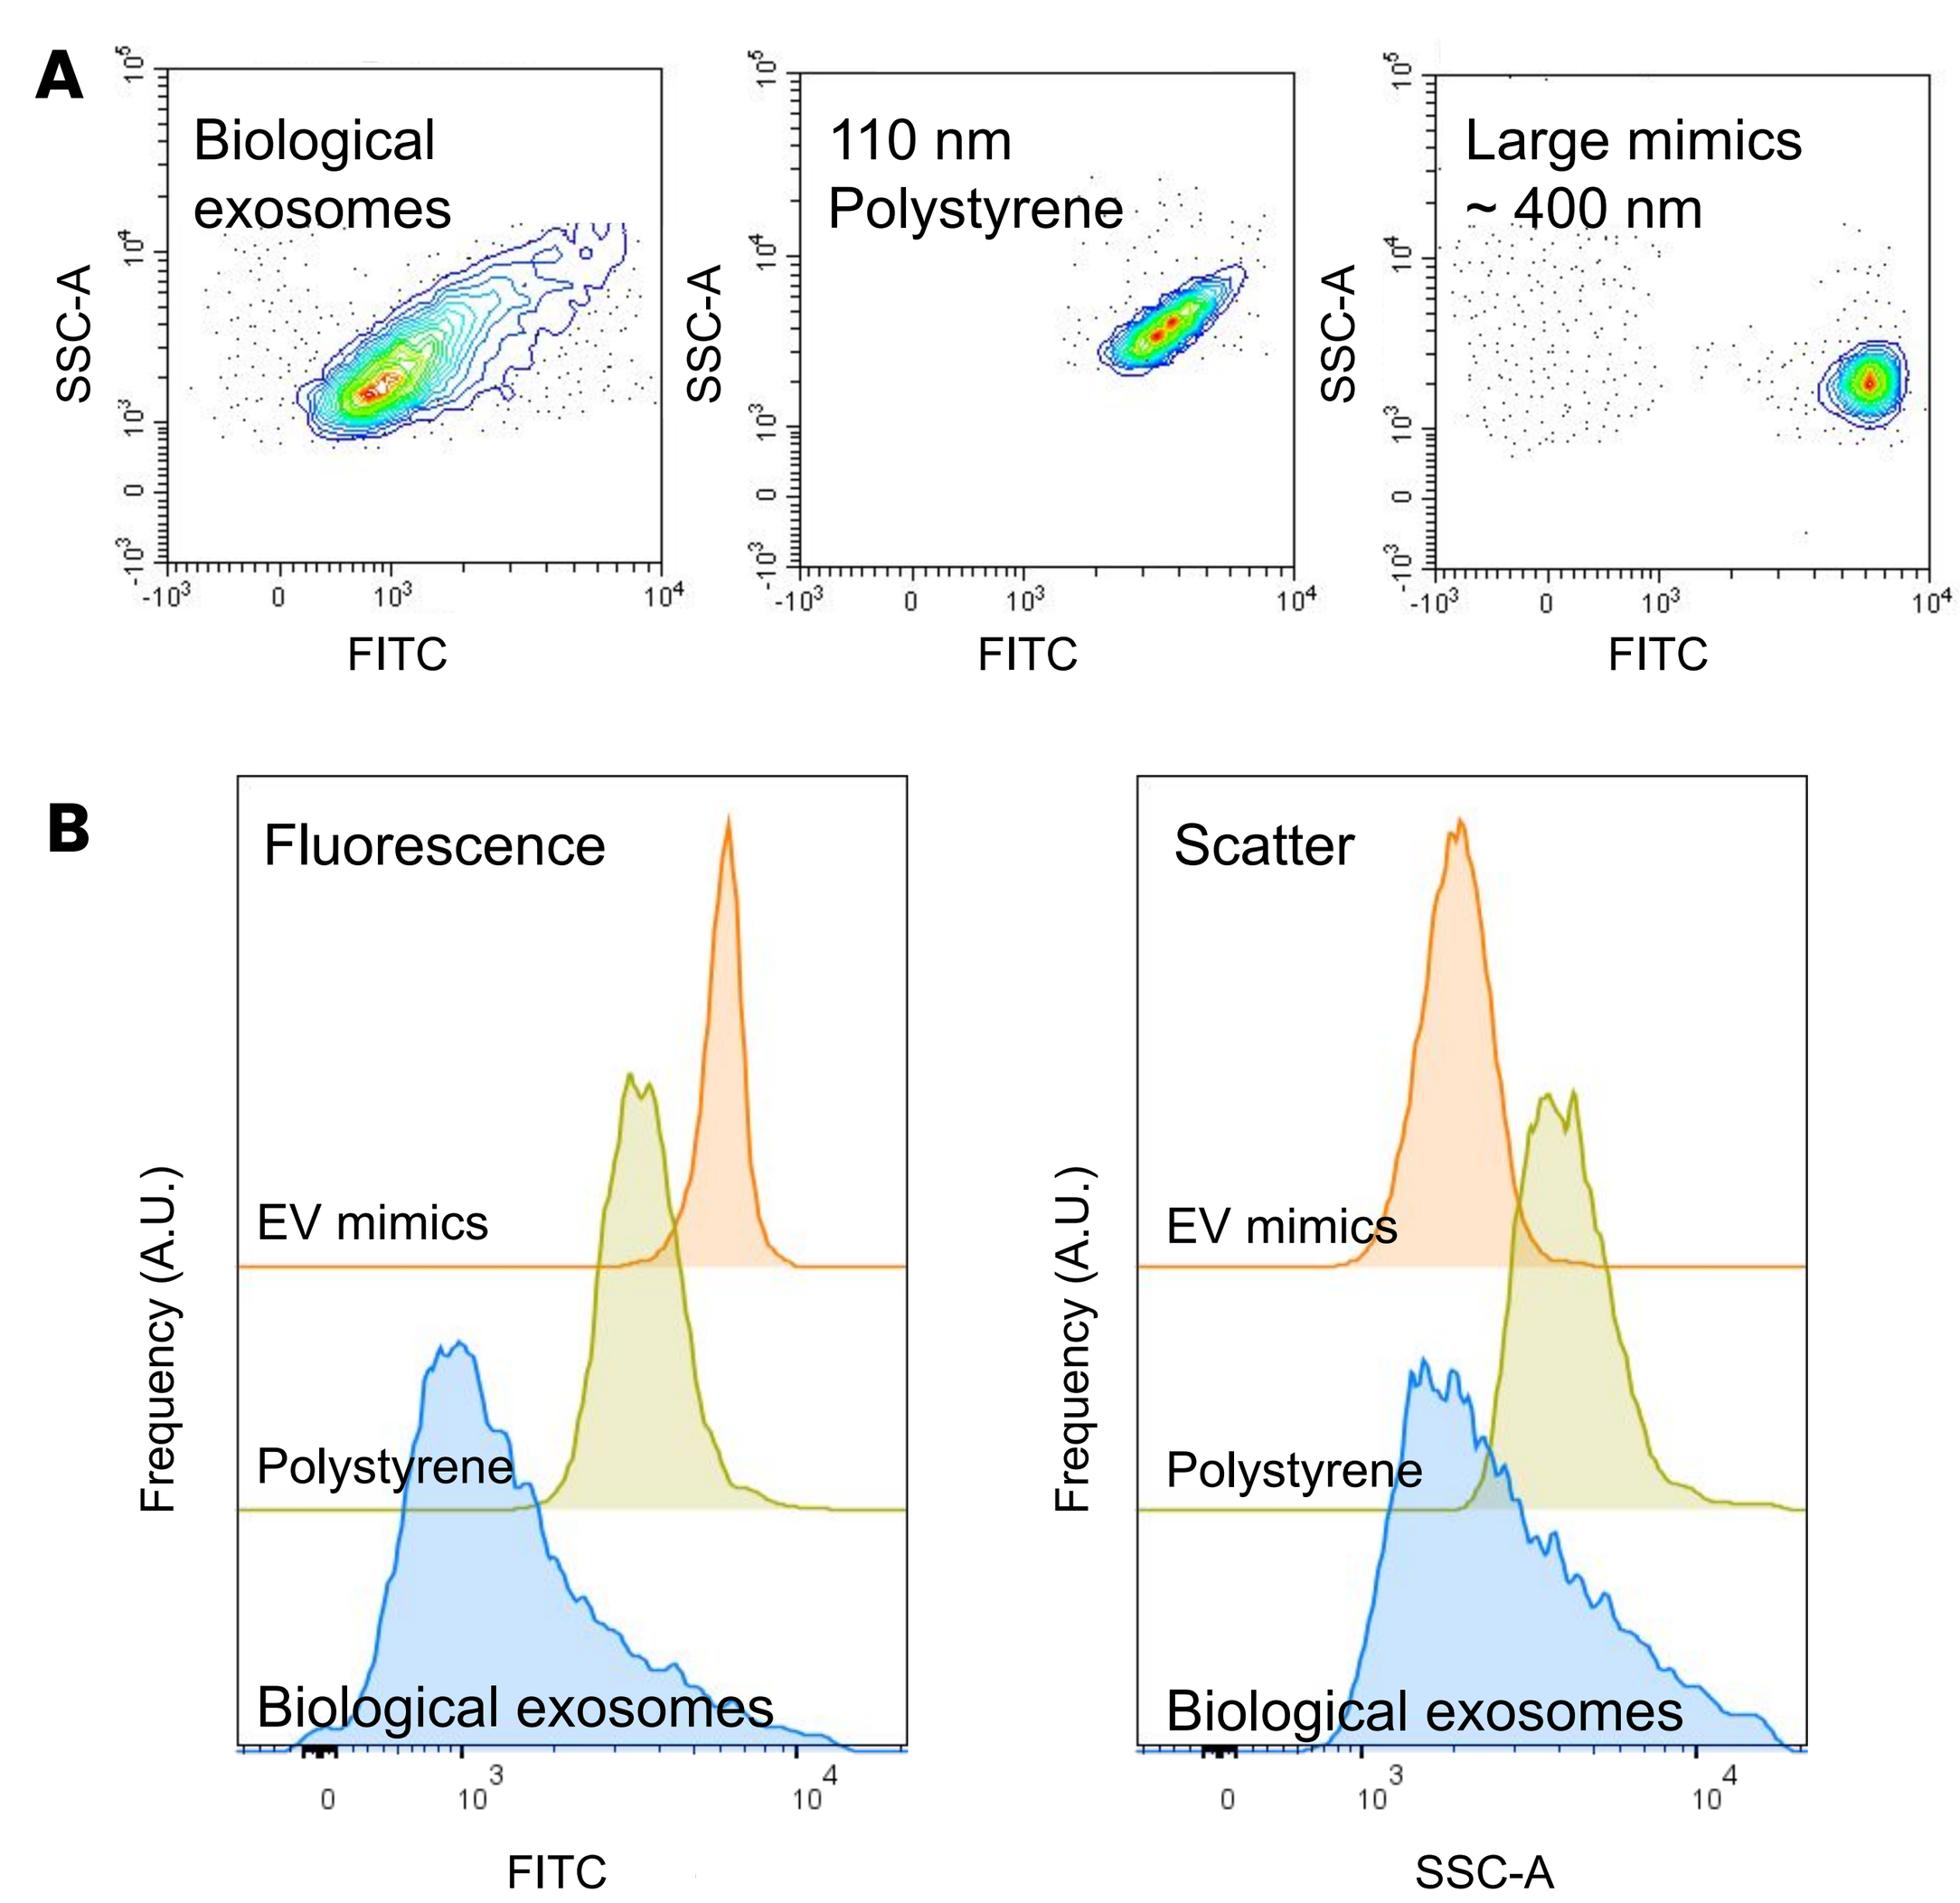
A
Biological exosomes
110 nm Polystyrene
Large mimics ~ 400 nm
SSC-A
FITC
B
Fluorescence
Scatter
EV mimics
Polystyrene
Biological exosomes
Frequency (A.U.)
FITC
SSC-A

Figure 2: **A.** Scatter profile and **B.** histograms comparing fluorescent labeled EV mimics with large exosomes (HansaBioMed) and polystyrene nanoparticles (Apogee). Slingshot EV mimics are monodisperse in both scatter and fluorescence, and feature EV-matching scatter intensity due to their lower RI.

Homogeneous Morphology

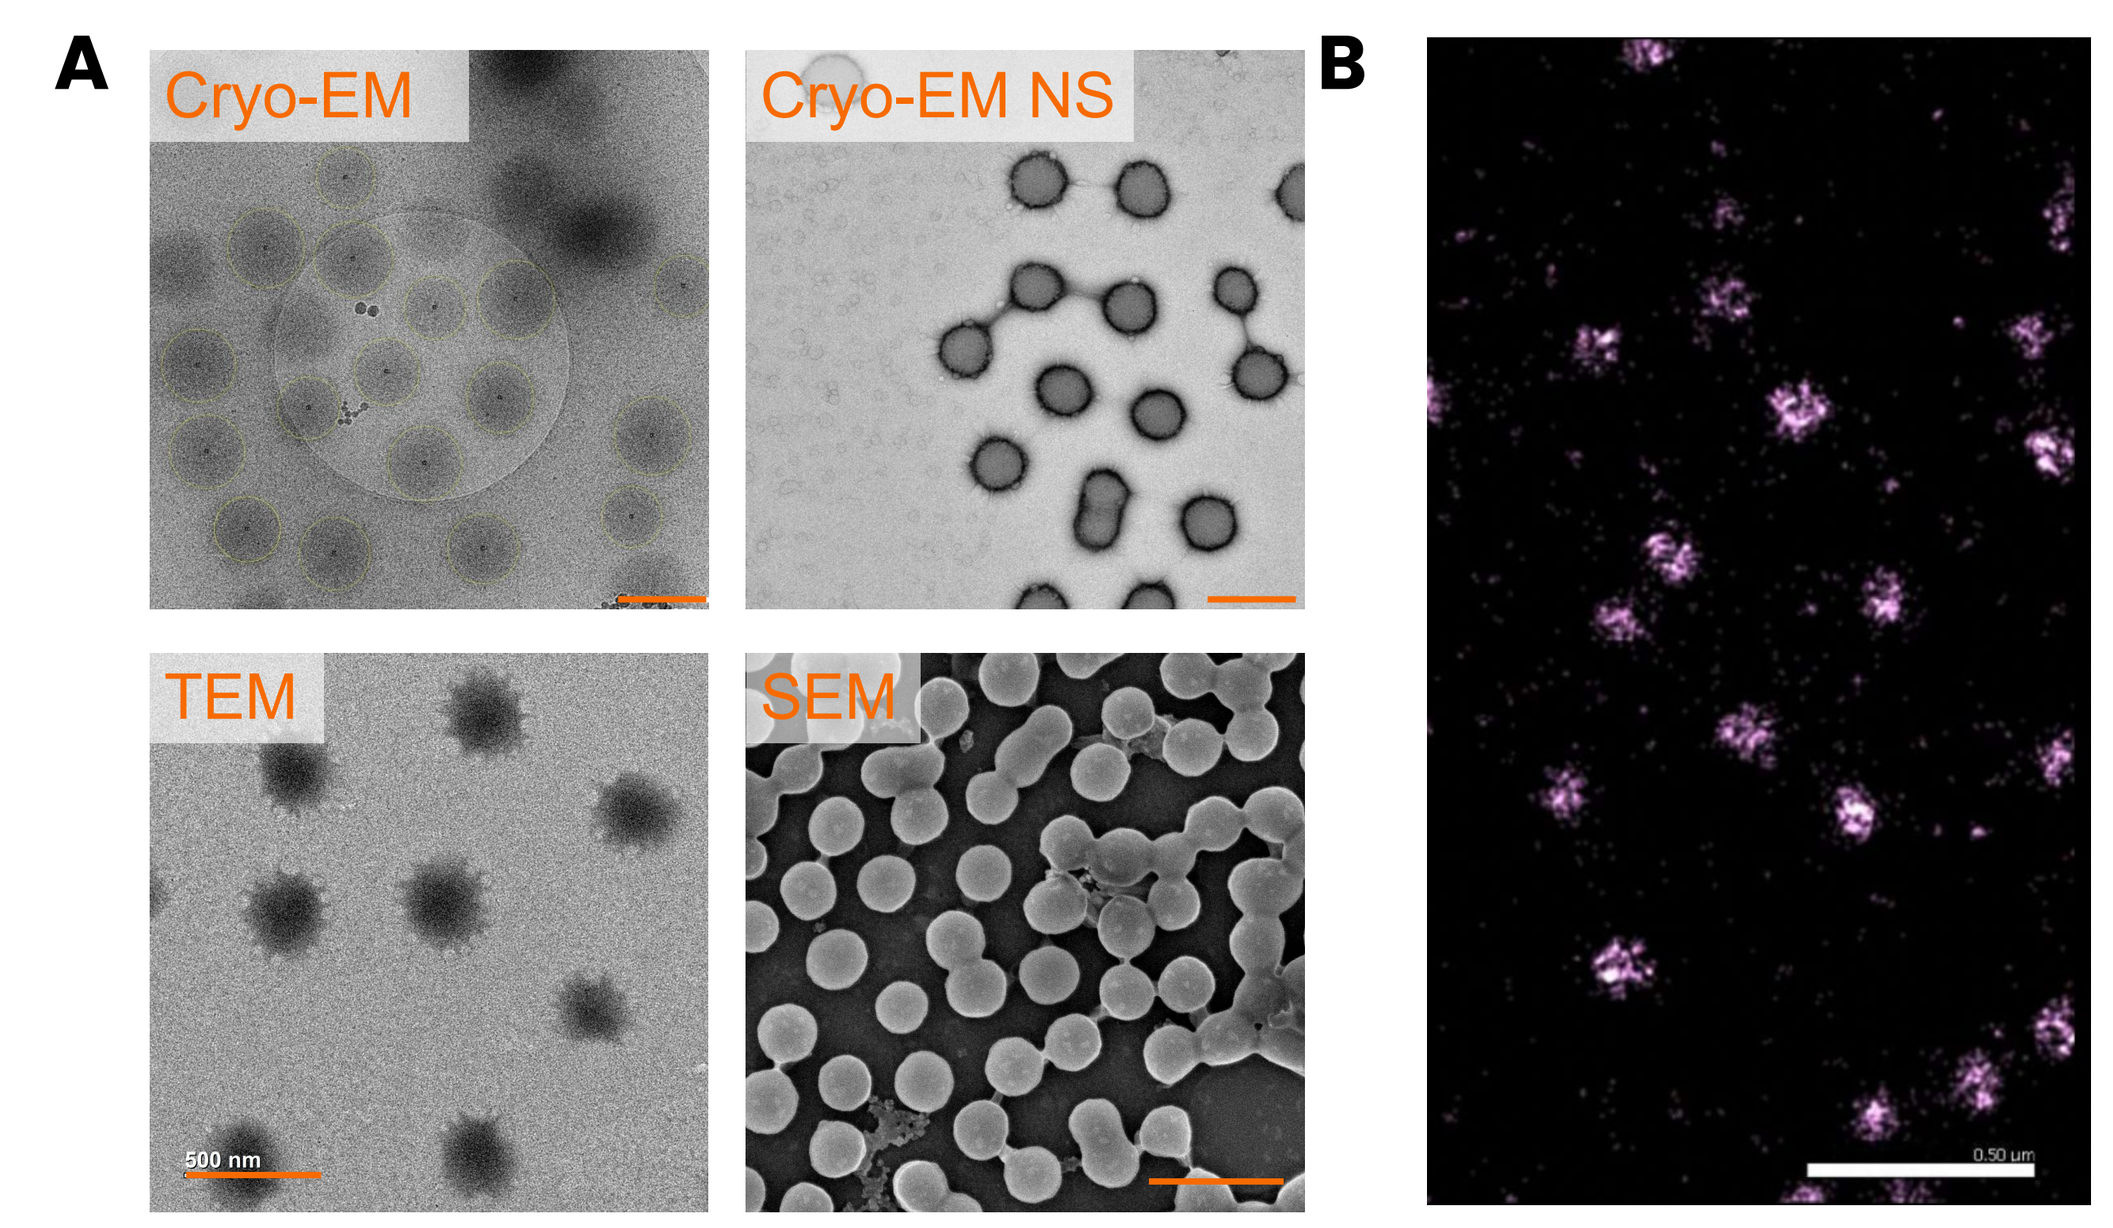
A
Cryo-EM
Cryo-EM NS
TEM
SEM
500 nm
B
0.50 µm

Figure 3: **A.** Morphology of large synthetic EV mimics, characterized by various electron microscopy methods. Cryo-EM represents the true size in an aqueous suspension, while other methods measures the dry size. **B.** Super-resolution microscopy of labeled medium EV mimics. Pseudo-color demonstrates the distribution of fluorophores. Scale bar = 500 nm.

Multi-level Fluorescence and Scatter

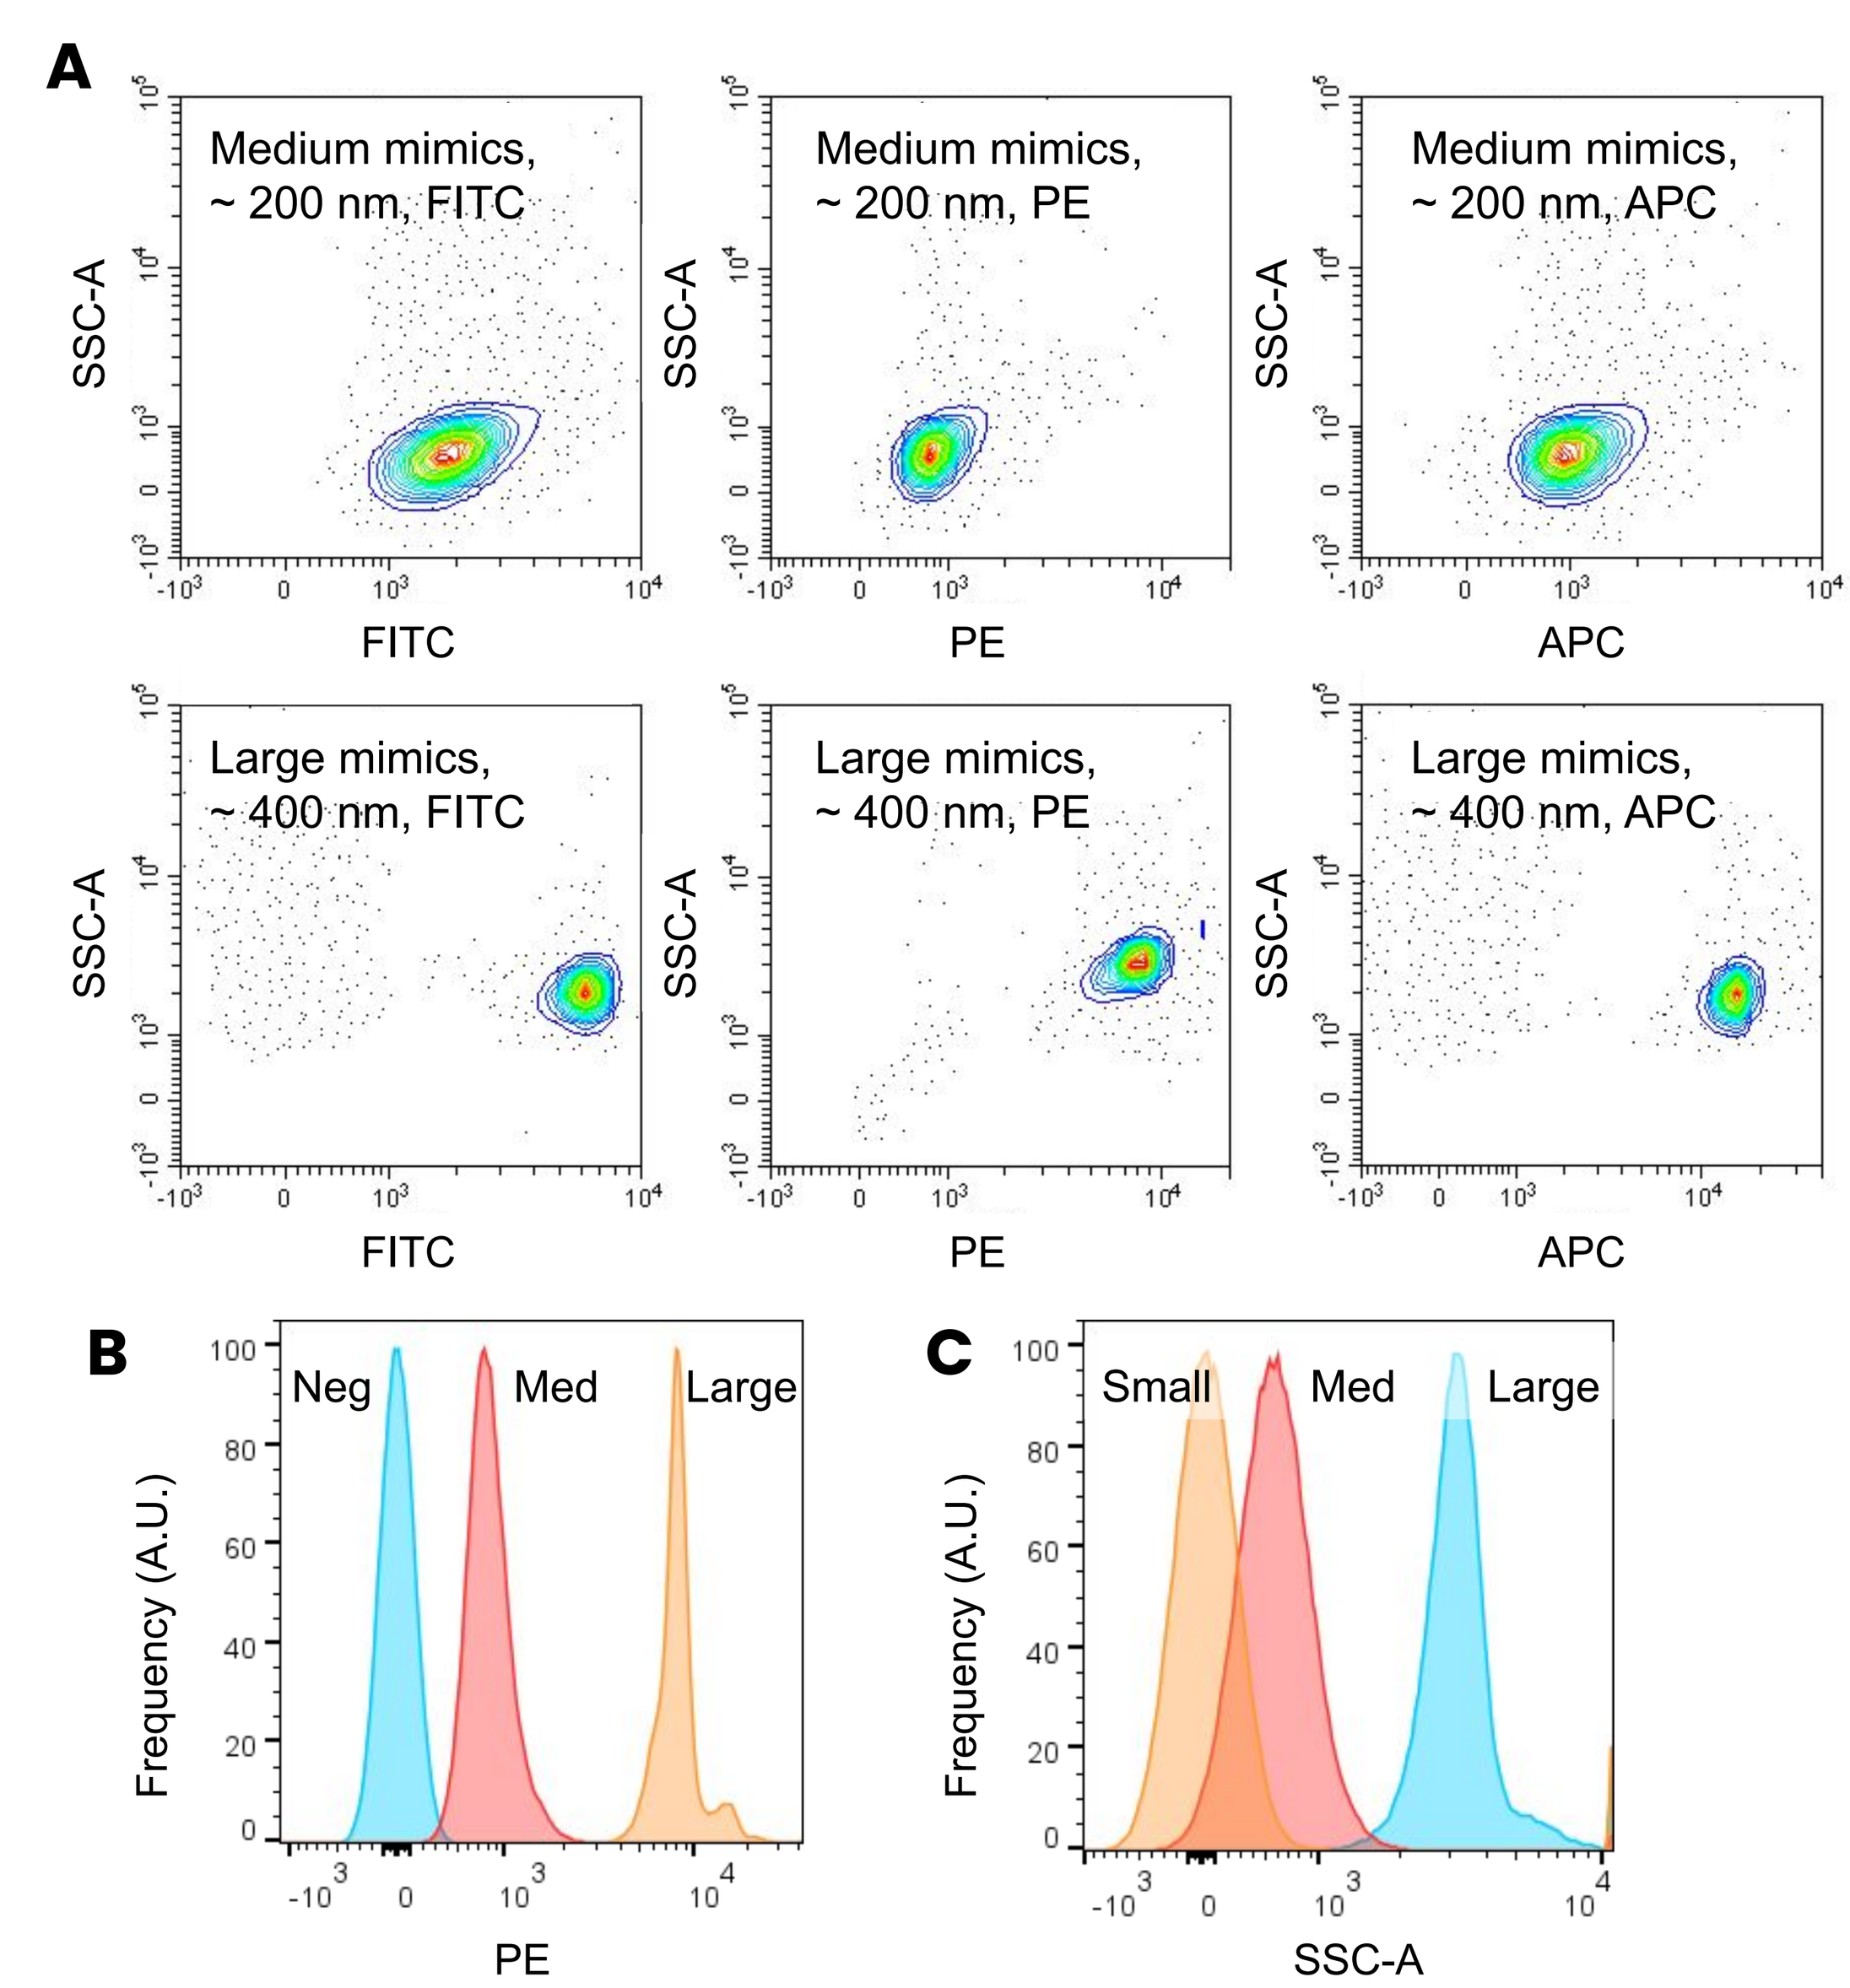
A
Medium mimics, ~ 200 nm, FITC
Medium mimics, ~ 200 nm, PE
Medium mimics, ~ 200 nm, APC
Large mimics, ~ 400 nm, FITC
Large mimics, ~ 400 nm, PE
Large mimics, ~ 400 nm, APC
SSC-A
FITC
PE
APC
B
Neg
Med
Large
Frequency (A.U.)
PE
C
Small
Med
Large
SSC-A

Figure 4: **A.** Various EV mimics species with different sizes and fluorescent labeling, size measured by DLS. **B.** A multi-level fluorescent reference in the PE channel consisting of APC-labeled medium mimics (negative) with PE-labeled medium and large mimics. **C.** A multi-level size estimation reference with PE-labeled EV mimics.

CONCLUSIONS

Slingshot Biosciences has developed polymer-based EV mimics with several key advantages:

- **Superior EV mimics that match key optical properties:** EV mimics demonstrate a lower refractive index (~1.38), and are more similar to biological EVs than current polystyrene or silica based nanoparticles. Additionally, both the size and refractive index can be independently customized for unique applications.
- **Well-defined fluorescence**: Despite the size limitation, EV mimics have well-defined fluorescence and low spillover or autofluorescence.
- **Ease of use:** EV mimics are non-hazardous and can be stored long-term without lyophilization. They also require minimal setup, eliminating the need for purification or reconstitution as seen with biological references.
- **Consistency and homogeneity:** EV mimics are more homogenous than current nanoparticles, both in terms of size and fluorescence. They are not sensitive to donor or processing variability which are common for biological references.

Slingshot Biosciences' EV mimics represent a promising advancement in flow cytometry standardization to support the growing demands of EV-based diagnostics and therapeutics. Potential applications include:

- **Assay validation:** The well-defined scatter and fluorescence profile of synthetic mimics enable easy optimization of EV flow cytometry assays against common pitfalls such as swarm detection and background noise.
- **Calibration:** EV mimics are superior as daily quality control material in maintaining consistency of EV flow cytometry measurements. The true-to-life scatter match allow accurate characterization of instrument shift at the correct scatter intensity level, which is not feasible with polystyrene beads.
- **Standardization and harmonization:** EV mimics offer superior logistics when deployed across instruments and lab sites. This standardization minimizes sources of variation that can confound experimental development and validation in longitudinal scientific research and mass clinical studies.

ACKNOWLEDGEMENTS

We would like to thank **Nanostics Inc.** for MALS RI measurement and **ONI Inc.** for super-resolution microscopy