Use of Custom TruCytes™ Cell Mimics as an Internal Assay Control to Validate a Prognostic Flow Cytometry Test Quantifying FcγRIIa Expression on Fixed Platelets

David J Schneider[1], Heidi S Taatjes-Sommer[1], Peter M DiBattiste[2], Kanwal S Palla[3], Tyler Shovah[3], Subhanip Biswas[3], Jeanne Ohrnberger[2] Annalise Barnette[3]

1. Department of Medicine, Cardiovascular Research Institute, The University of Vermont, Burlington, VT, 05401, USA.
2. Prolocor Inc, Philadelphia, PA, 19146, USA.
3. Slingshot Biosciences Inc, Emeryville, CA, 94608, USA.

ABSTRACT

Background:
The FcγRIIa receptor plays a crucial role in platelet activation, which is linked to thrombosis—a leading cause of heart attack and stroke. Platelet FcγRIIa (pFCG) is elevated in patients with high cardiovascular risk, making it a useful prognostic marker. The Prolocor pFCG® test is an advanced flow cytometry-based assay that accurately quantifies FcγRIIa expression on fixed platelets, helping clinicians assess thrombosis risk and make informed treatment decisions. The use of Slingshot Biosciences' TruCytes™ cell mimics as an internal assay control improves precision, accuracy, and linearity across a broad range of biological expression.

Methods:
The pFCG® test uses a novel 5G1 antibody to bind FcγRIIa on fixed platelets, which limits platelet degradation and enhances assay precision. Validation studies assessed assay performance by comparing the known anti-FcγRIIa antibody (FL18.26) with the 5G1 antibody in the pFCG® test. Accuracy and precision of the test was evaluated with biological specimens (platelets) and custom-engineered TruCytes™cell mimics decorated with FcγRIIa.

Results:
The 5G1 antibody exhibited higher affinity for FcγRIIa than FL18.26, able to identify ~300 more molecules per platelet compared to FL18.26, as quantified by flow cytometry. Assay validation with biological specimens showed an intra-assay CV of 2.1 ± 0.1% and inter-assay CVs of 4.5 ± 1% (intraday) and 6.5 ± 0.4% (up to 5 days post-fixation). Linearity of the pFCG® test was demonstrated using TruCytes™cell mimics (R^2 = 0.984) over a range of expression. Inter-assay CVs were 2% ± 0.6% (intraday) and 9.9% ± 2.1% (interday across 20 non-consecutive days), well below the FDA guidance (25%) for acceptable variability. The extended stability of the TruCytes™cell mimics compared to the biological samples supports their use as reliable internal controls for the pFCG® test.

Conclusion:
The Prolocor pFCG® test is a highly precise and consistent prognostic tool for assessing thrombosis risk. Quantifying platelet reactivity using fixed platelets allows for broader application of this test, and the use of TruCytes™cell mimics as an internal control ensures precise and accurate FcγRIIa quantification to guide individualized treatment selection in clinical settings.

Evaluation of pFCG® assay precision

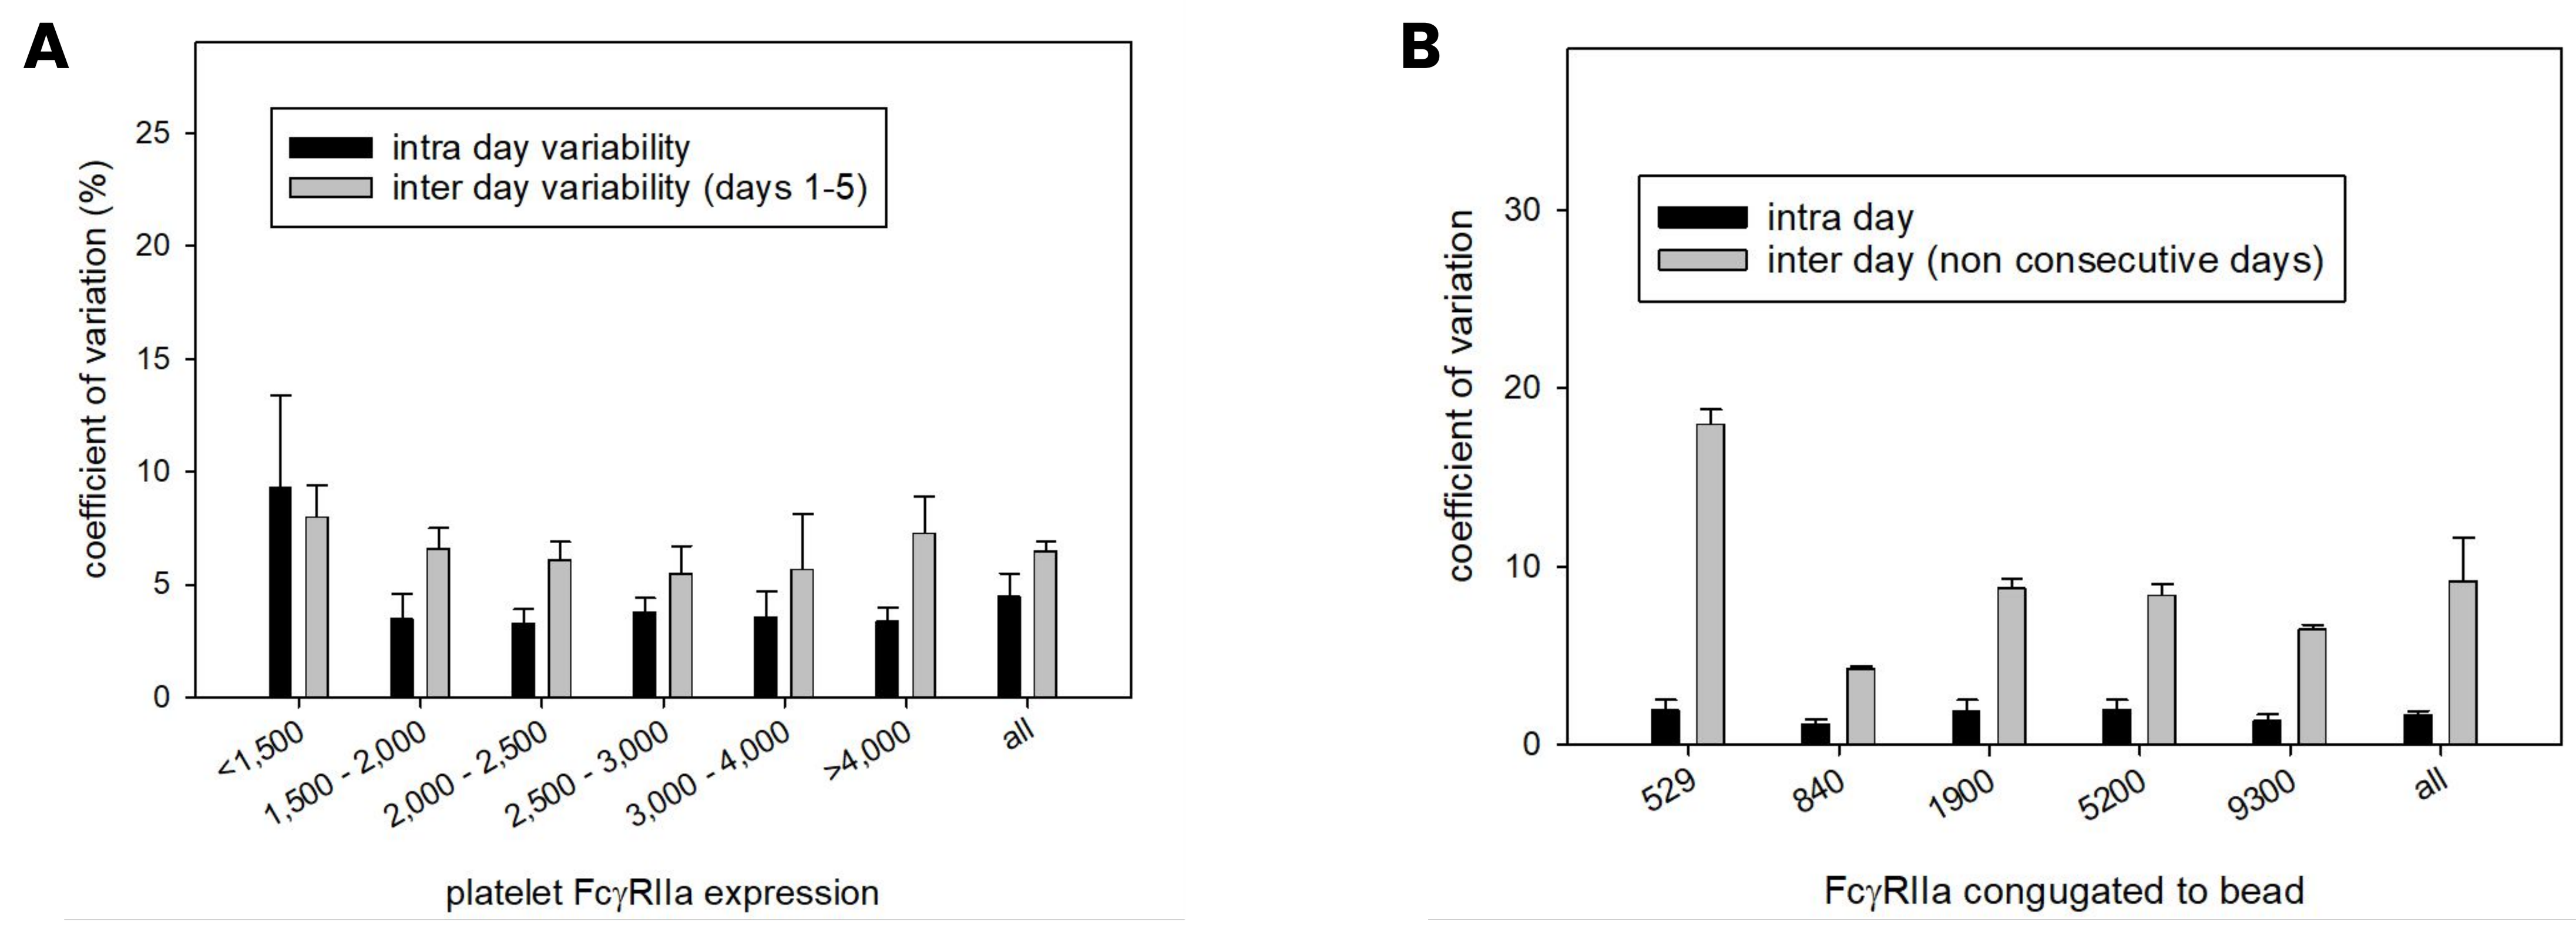
A
intra day variability
inter day variability (days 1-5)
coefficient of variation (%)
0
5
10
15
20
25
<1,500
1,500 - 2,000
2,000 - 2,500
2,500 - 3,000
3,000 - 4,000
>4,000
all
platelet FcγRIIa expression
B
intra day
inter day (non consecutive days)
coefficient of variation
0
10
20
30
529
840
1900
5200
9300
all
FcγRIIa congugated to bead

Figure 1. Assessment of assay precision. A) The interassay CV assessed across 50 biological samples. Intraday samples were performed on the same day with the second pFCG™ test performed at least 2h after the first test. Interday samples were performed on days 1–5 after fixation of samples. B) The interassay CV assessed with FcγRIIa-conjugated TruCytes™. Intraday samples were performed on the same day with the second pFCG™ test performed at least 2h after the first test. Interday samples were performed on 20 non-consecutive days.

Evaluation of FcγRIIa quantitation and pFCG® assay linearity

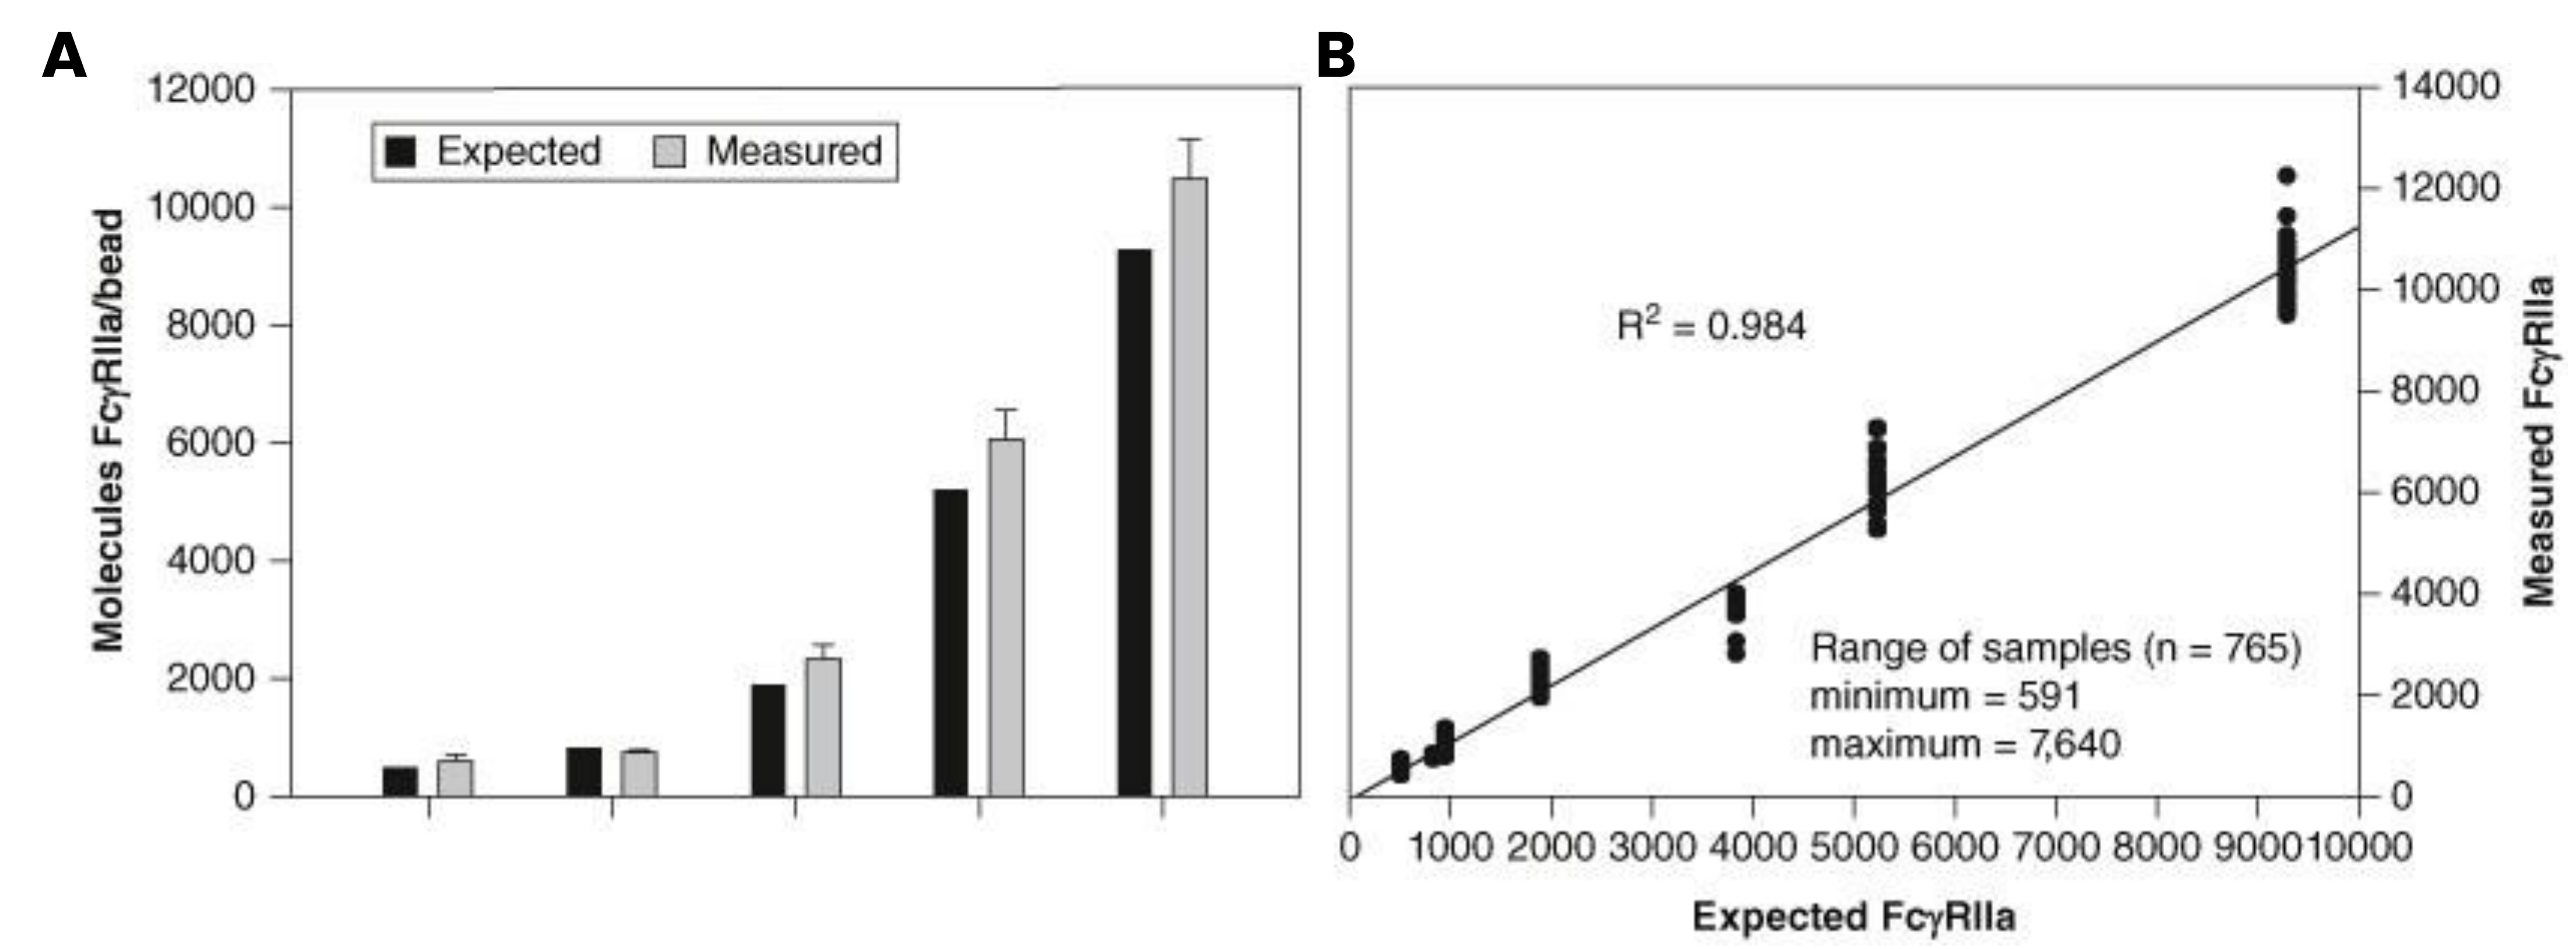
A
Expected
Measured
Molecules FcγRIIa/bead
0
2000
4000
6000
8000
10000
12000
B
R² = 0.984
Range of samples (n = 765)
minimum = 591
maximum = 7,640
0
1000 2000 3000 4000 5000 6000 7000 8000 9000 10000
Expected FcγRIIa
0
2000
4000
6000
8000
10000
12000
14000
Measured FcγRIIa

Figure 2. Evaluation of FcγRIIa quantitation and pFCG® assay linearity. A) The pFCG test accurately quantified FcγRIIa protein expression on the TruCytes™. Results were compared with the expected results. B) Linearity of the pFCG™ test was assessed with the TruCytes™. The pFCG™ test was performed (n = 765 determinations) on samples that ranged from 591 to 7640 molecules of FcγRIIa/cell mimic.

Assessment of pFCG® Intra-Assay Coefficient of Variation

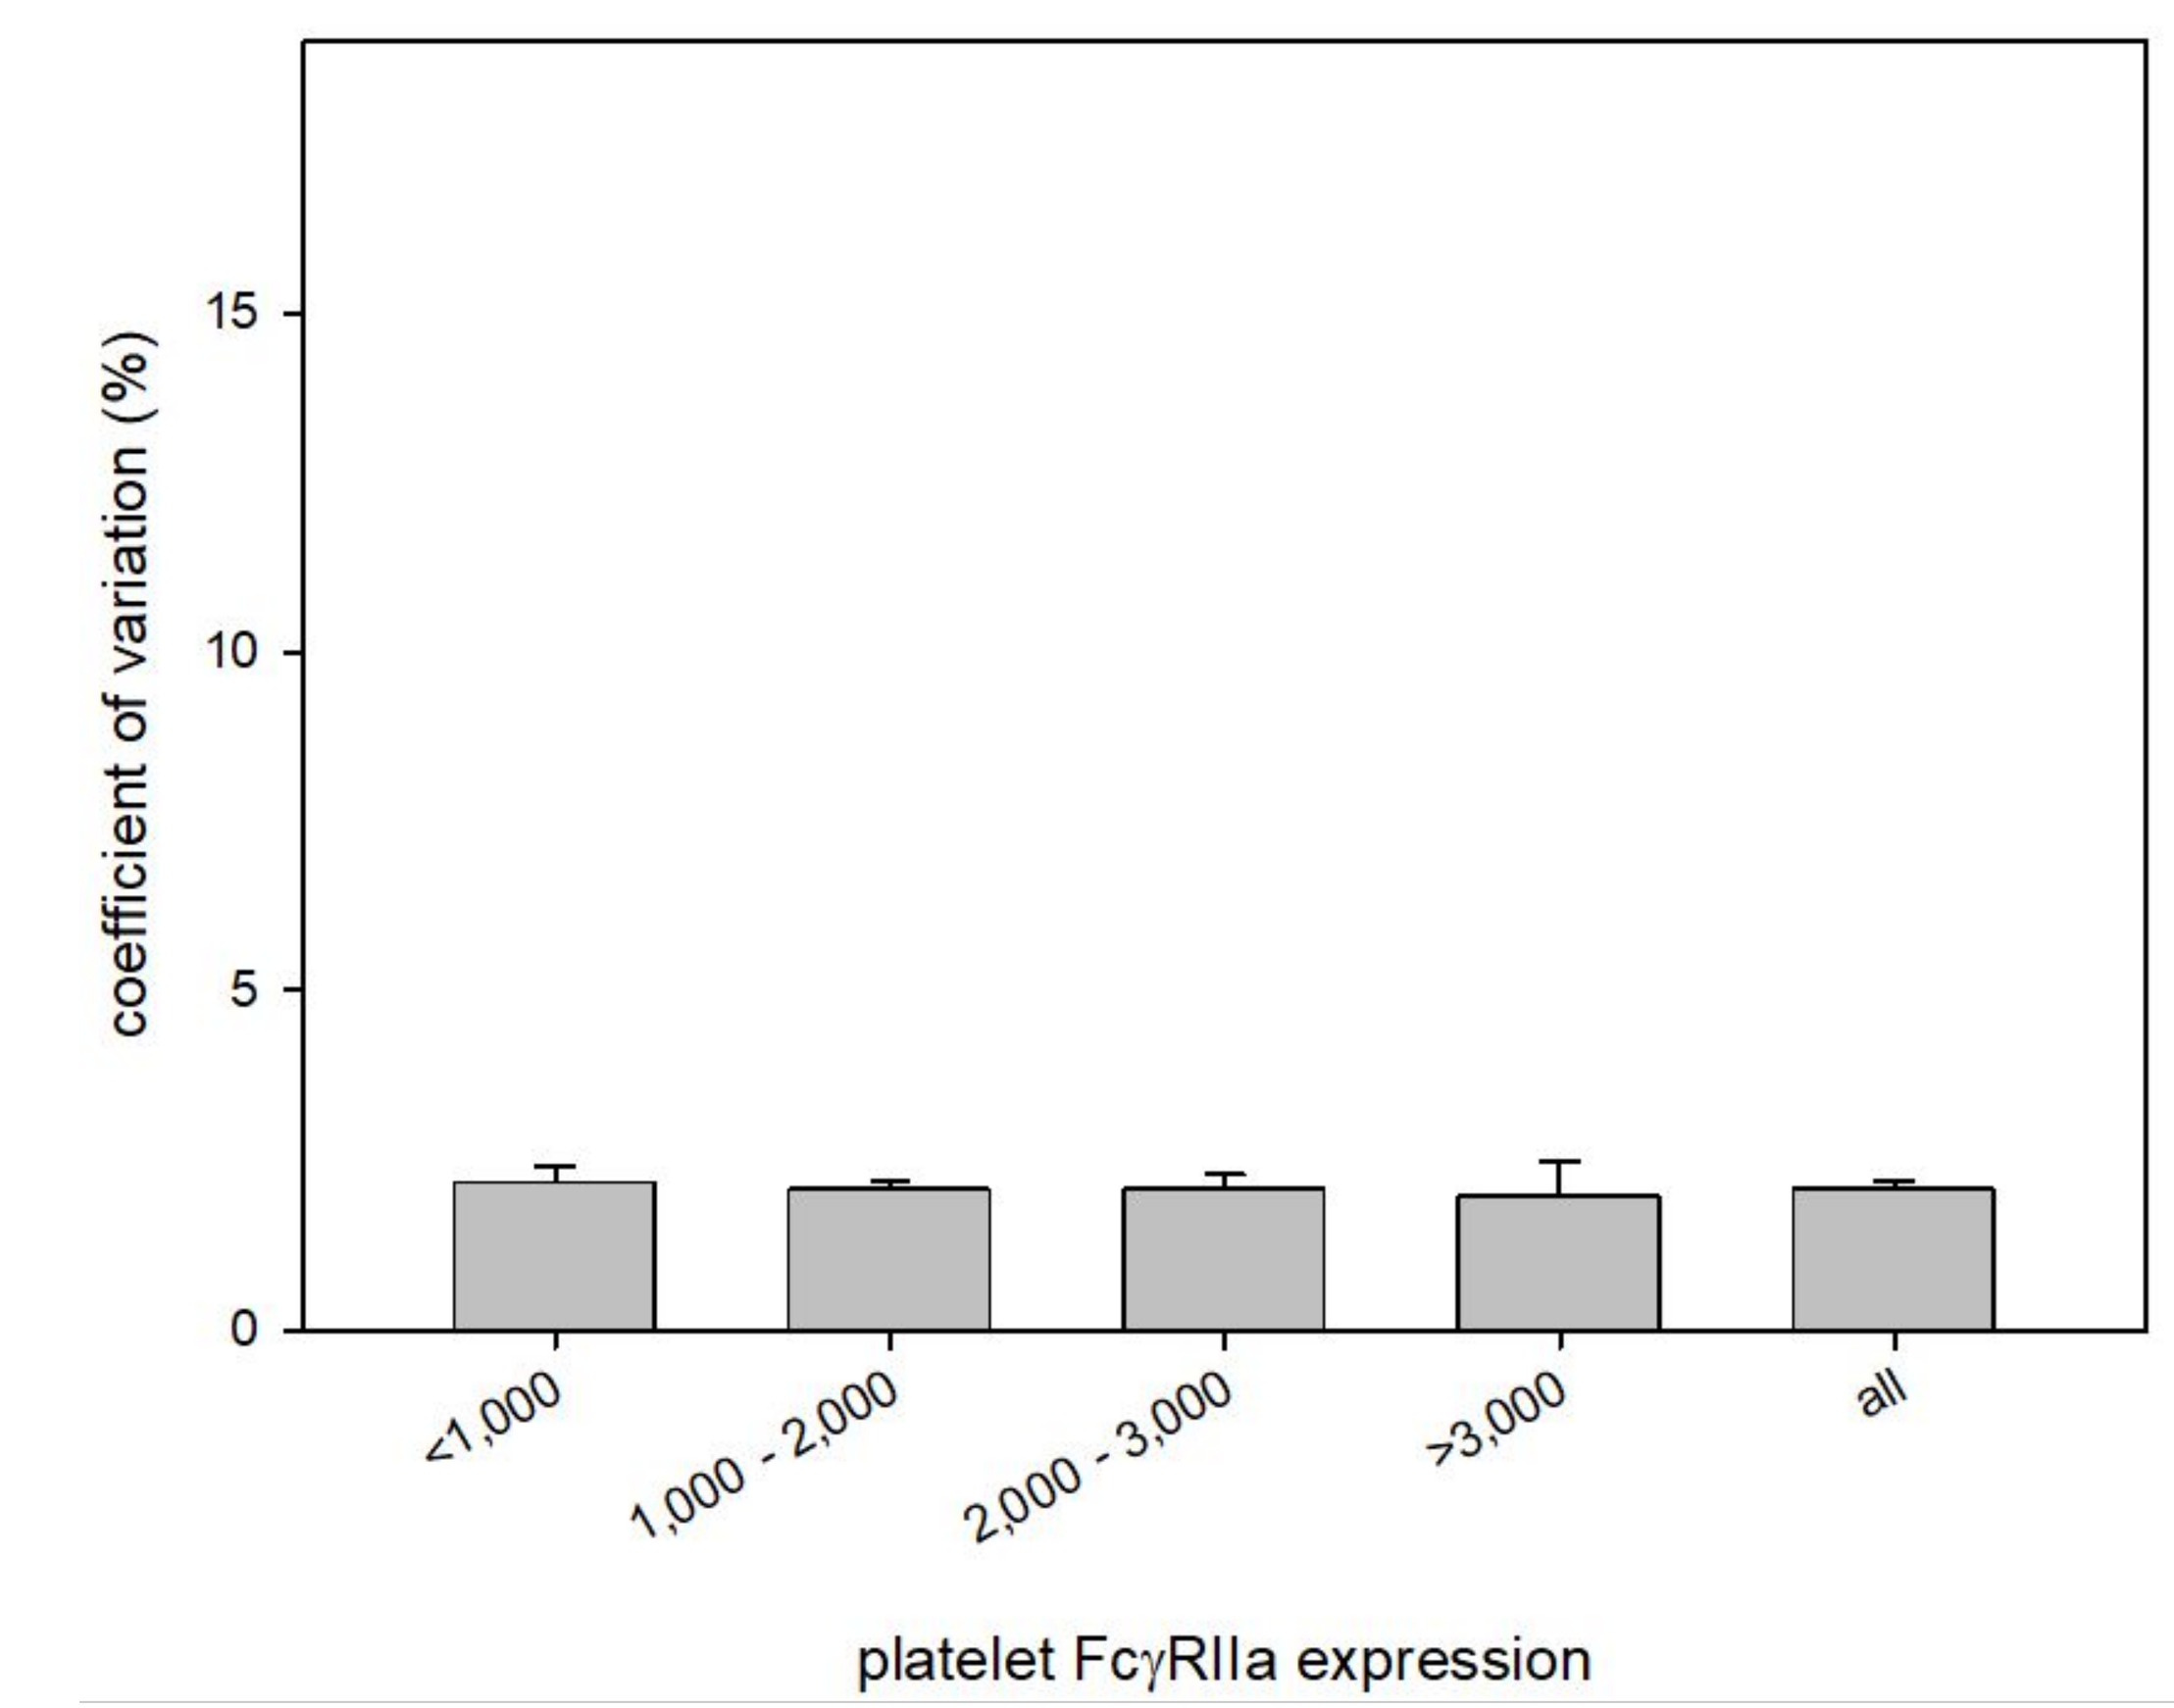
coefficient of variation (%)
0
5
10
15
<1,000
1,000 - 2,000
2,000 - 3,000
>3,000
all
platelet FcγRIIa expression

Figure 3. The intra-assay CV was assessed during the performance of the assay on biological specimens (platelets from 750 patients).
CV: Coefficient of variation.

SUMMARY

Slingshot engineers TruCytes™ cell mimics that express unique diagnostic markers such as FcγRIIa that aid in the assessment of new diagnostic assays.

- TruCytes™ allowed for the assessment of interassay precision, showing consistent results across multiple tests performed on different days. The coefficient of variation (CV) for the TruCytes™ was comparable to that observed with biological specimens, indicating that the test is reliable when using either sample type.
- Analytical validation studies demonstrate the test's high accuracy, with intra-assay variation at just 2% and inter-assay variation at 10%, both well below the FDA's acceptable threshold of 25%.
- TruCytes™cell mimics demonstrated extended stability compared to biological samples (closed vial stability at -20◦C of 18 months. Reconstituted stability of 30 days at 4◦C.)