Synthetic CD34+ Stem Cell Mimics as a Stable and Reproducible Control for Flow Cytometry

Swetha Gunturu, Subhanip Biswas, Valerie Chen, Brian Kerfs, William Pratcher, Kanwal Palla

Slingshot Biosciences, Inc

ABSTRACT

Background:
CD34⁺ hematopoietic stem cells are routinely measured by flow cytometry for clinical and research applications. Reliable control materials are important for instrument setup, assay development, quality control, and consistent data analysis. Biological controls often face variability, stability and handling limitations. A standardized synthetic control material provides an alternative approach for supporting CD34 flow cytometry workflows.

Methods:
Synthetic CD34⁺ Cell Mimics (Slingshot Bio) were evaluated using the ISHAGE CD34 enumeration strategy on conventional and spectral flow cytometers. The control, containing defined CD45⁺CD34⁺, CD45⁺ only, and dead cell mimic populations was examined for compatibility to top antibody clones and comparison against commercial CD34 control materials. Intermediate precision was assessed across multiple operators, instruments, and days. Spike-in studies were completed in bone marrow and stabilized whole blood samples. Accelerated closed-vial stability testing utilized Arrhenius modeling.

Results:
CD34⁺ Cell Mimics demonstrated clear CD45⁺CD34⁺ resolution using the ISHAGE CD34 enumeration strategy across all cytometers, with staining comparable to commercial control materials. Intermediate precision testing across multiple operators, instruments, and days demonstrated less than 5% CV for all cell mimic populations. Spike-in studies supported recovery and detection of CD34⁺ populations in bone marrow and stabilized whole blood samples. Closed-vial stability was equivalent to 24 months.

Conclusions:
CD34⁺ Cell Mimics demonstrated consistent performance across flow cytometry platforms, operators, and testing conditions. The demonstrated precision, stability, antibody clone compatibility, and recovery in spiked samples support their potential use as a standardized control material for CD34 flow cytometry quality control and assay evaluation.

PRODUCT INFORMATION

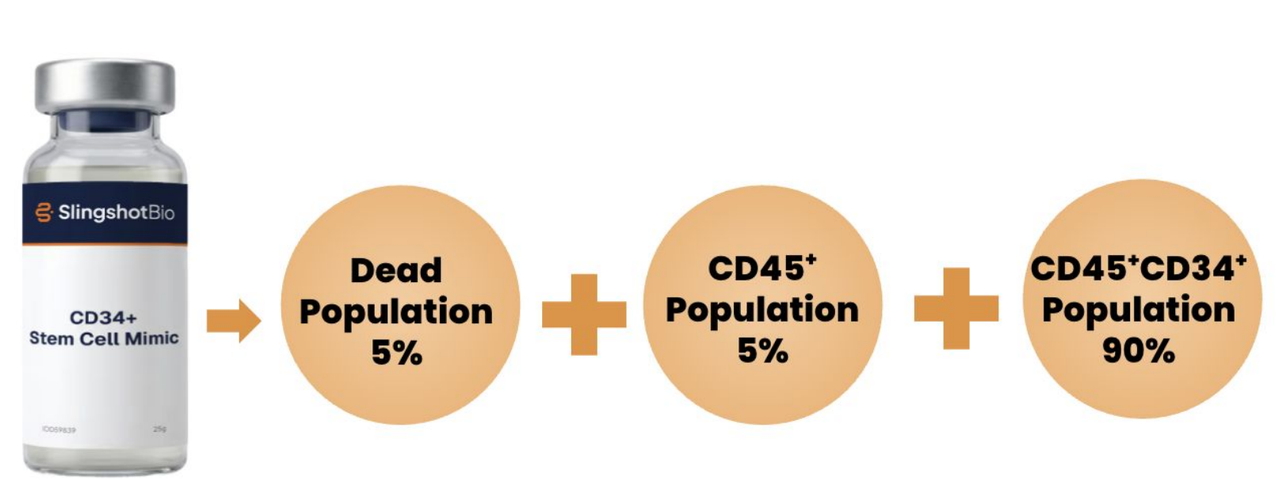

SlingshotBio
CD34+ Stem Cell Mimic
Dead Population 5%
CD45⁺ Population 5%
CD45⁺CD34⁺ Population 90%

Biomarker	Clone/Reagent Compatibility		
CD45	2D1	HI-30	J33
CD34	8G12	561	4H11
Viability	Compatible with DNA Viability dyes		

CD34⁺ Stem Cell Mimic™ contains CD34, CD45, and viability (DNA) markers with lymphocyte-like scatter properties and a high-density CD34⁺ population (~90% of CD45⁺ cells). It is a lyophilized product stored at −20 °C.

RESULTS: CD34+ Stem Cell Mimics Compatible with ISHAGE gating strategy

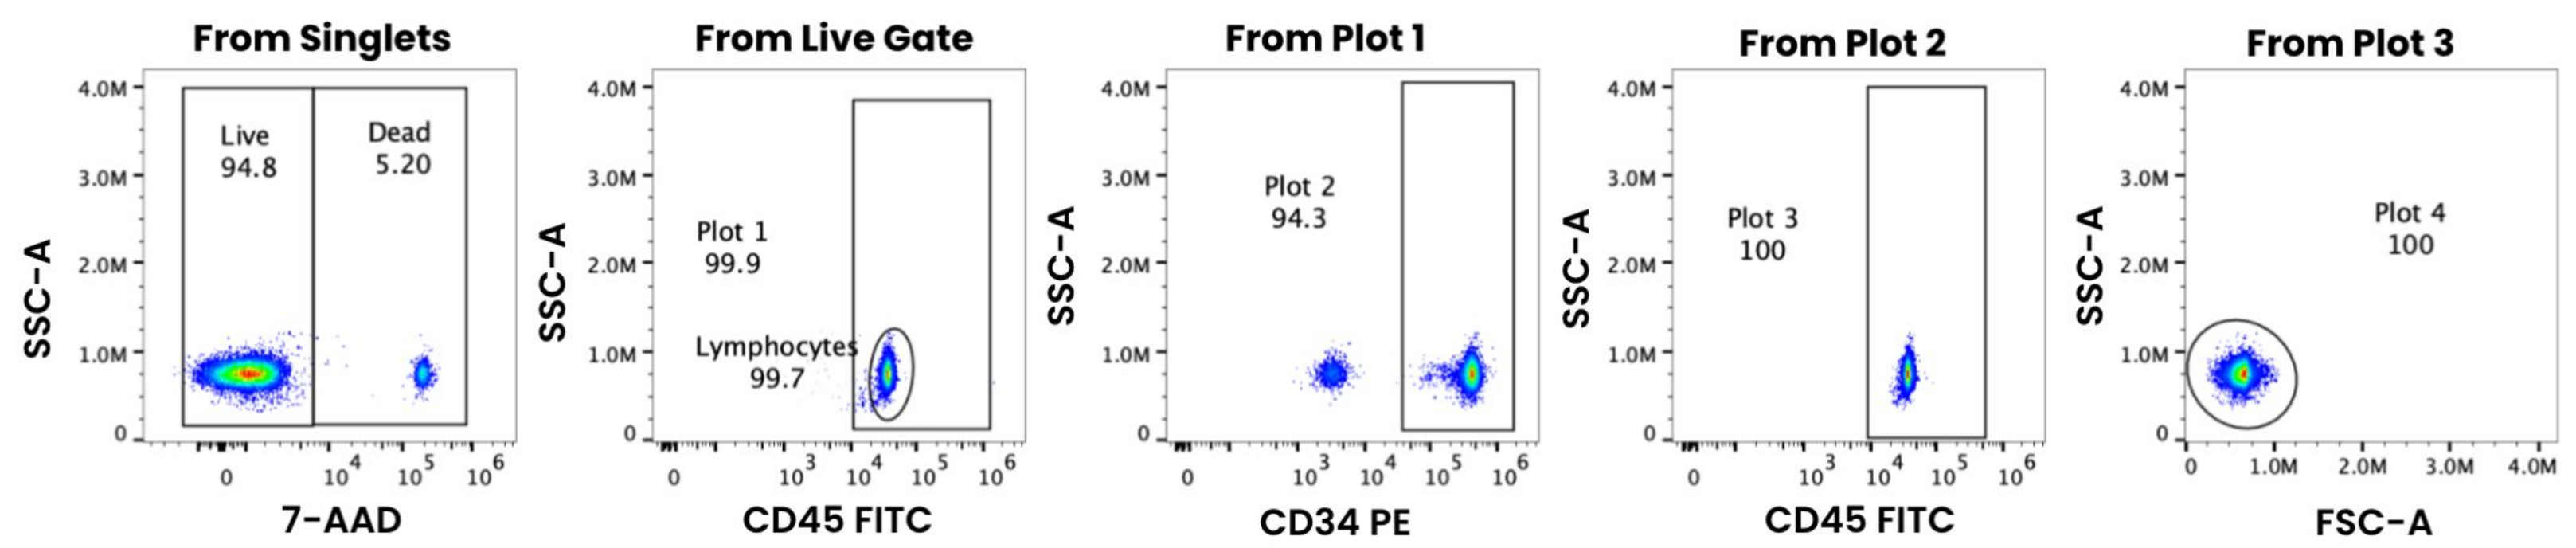

From Singlets
Live 94.8
Dead 5.20
7-AAD
From Live Gate
Plot 1 99.9
Lymphocytes 99.7
CD45 FITC
From Plot 1
Plot 2 94.3
CD34 PE
From Plot 2
Plot 3 100
CD45 FITC
From Plot 3
Plot 4 100
FSC-A
SSC-A

Figure 1. Representative flow cytometry plots showing the identification of CD34 Stem Cell Mimics using the ISHAGE gating strategy. The CD34-positive population was clearly resolved through sequential gating.

RESULTS: CD34+ Stem Cell Mimics demonstrate high Reproducibility for Intermediate Precision (Within Laboratory)

Population Type	%CV between three replicates when tested on same day on same instrument	%CV when same operator tested on three different days	%CV when same operator tested on three different instruments on same day	%CV when two different operators tested on same instrument on same day
CD45+CD34+	0.12	0.17	0.34	0.07
CD45+ Only	1.96	1.63	4.40	3.52
Dead (7-AAD)	1.47	1.48	0.90	1.34

Table 1. Intermediate precision (%CV) of CD34 Stem Cell Mimics across multiple operators, instruments, and days for CD45+CD34+, CD45+ only, and dead cell populations.

RESULTS: CD34+ Stem Cell Mimics demonstrated highly stability in closed vial storage for up to 24 months

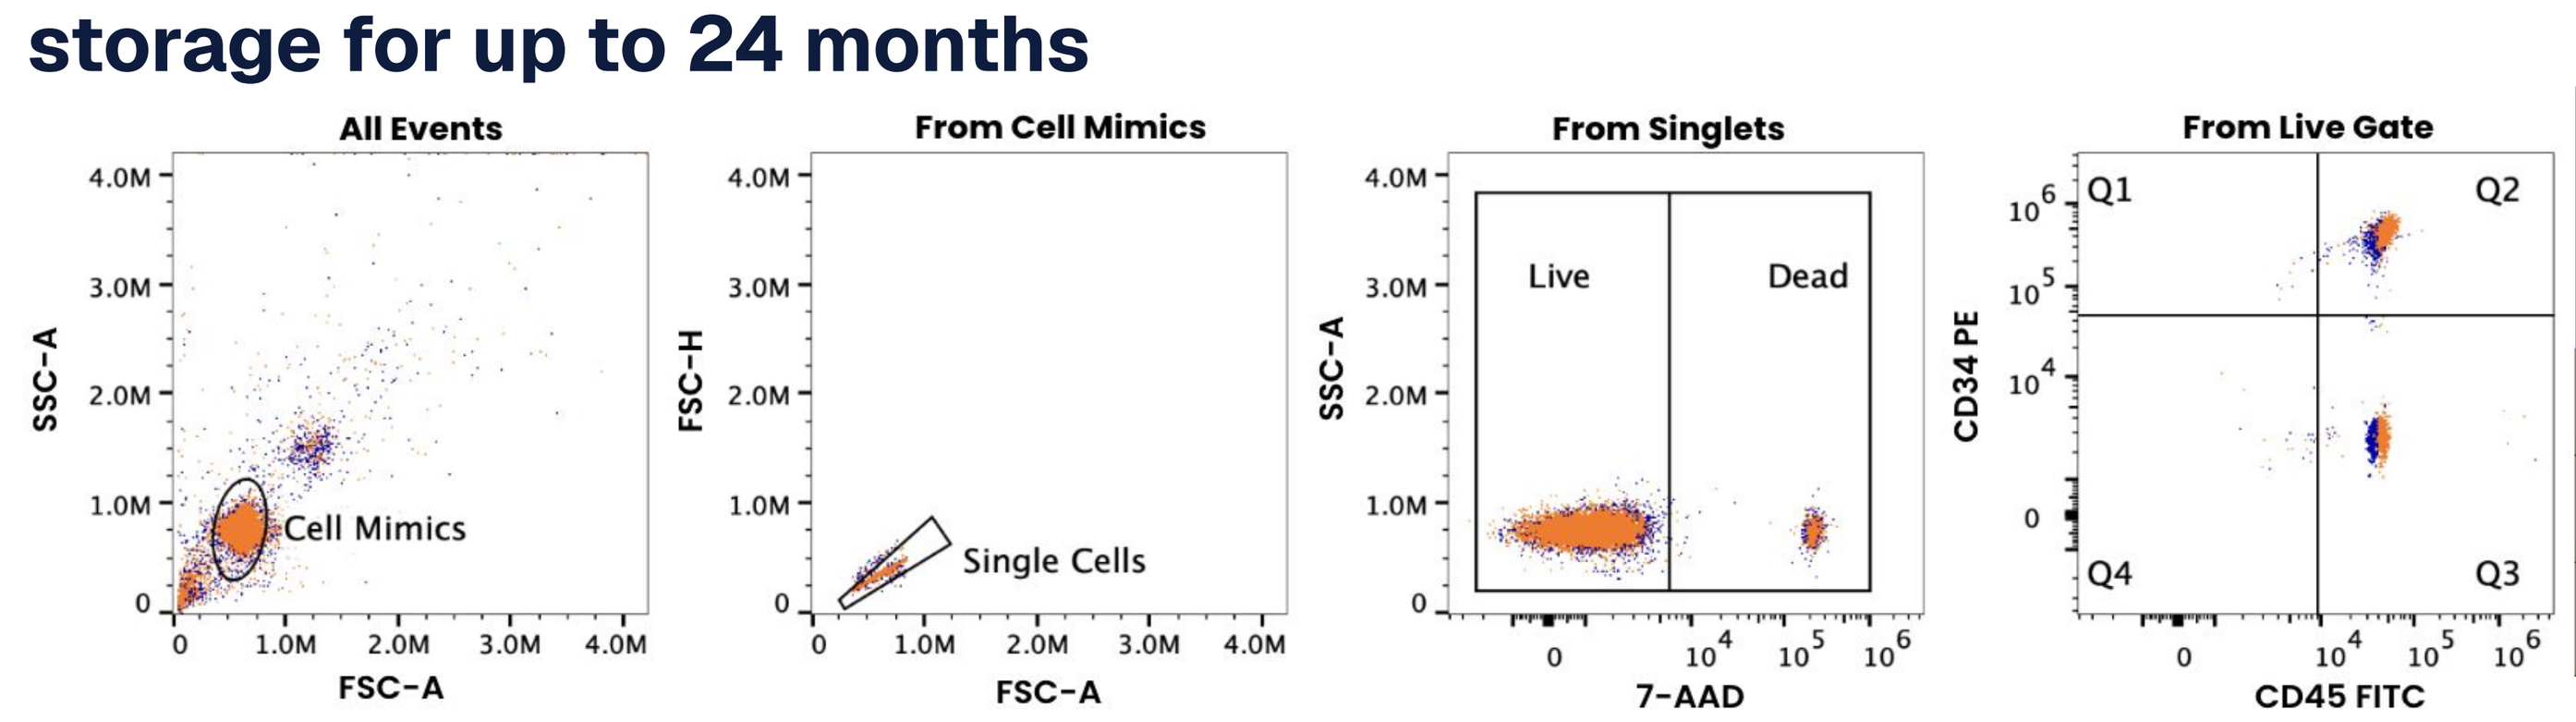

All Events
Cell Mimics
FSC-A
SSC-A
From Cell Mimics
Single Cells
FSC-H
From Singlets
Live
Dead
7-AAD
From Live Gate
Q1
Q2
Q3
Q4
CD34 PE
CD45 FITC

Population Type	Cumulative %CV of all timepoints	Absolute difference between T0 and T24
CD45+CD34+	0.16	-0.24
CD45+ Only	2.23	2.96
Dead (7-AAD)	1.15	0.33

Table 2. (Left) Stability of CD34⁺ Stem Cell Mimics across timepoints, assessed under accelerated conditions at 37 °C using the Arrhenius model. Cumulative %CV across all timepoints and absolute difference between T0 and T24 were evaluated for CD45+CD34+, CD45+ only, and dead cell populations. Figure 2. (Right) Dotplot overlay of Timepoints T0 (Orange) and T24 (Blue)

RESULTS: CD34 Stem Cell Mimics Demonstrated Compatibility with Reagents from Multiple Vendors

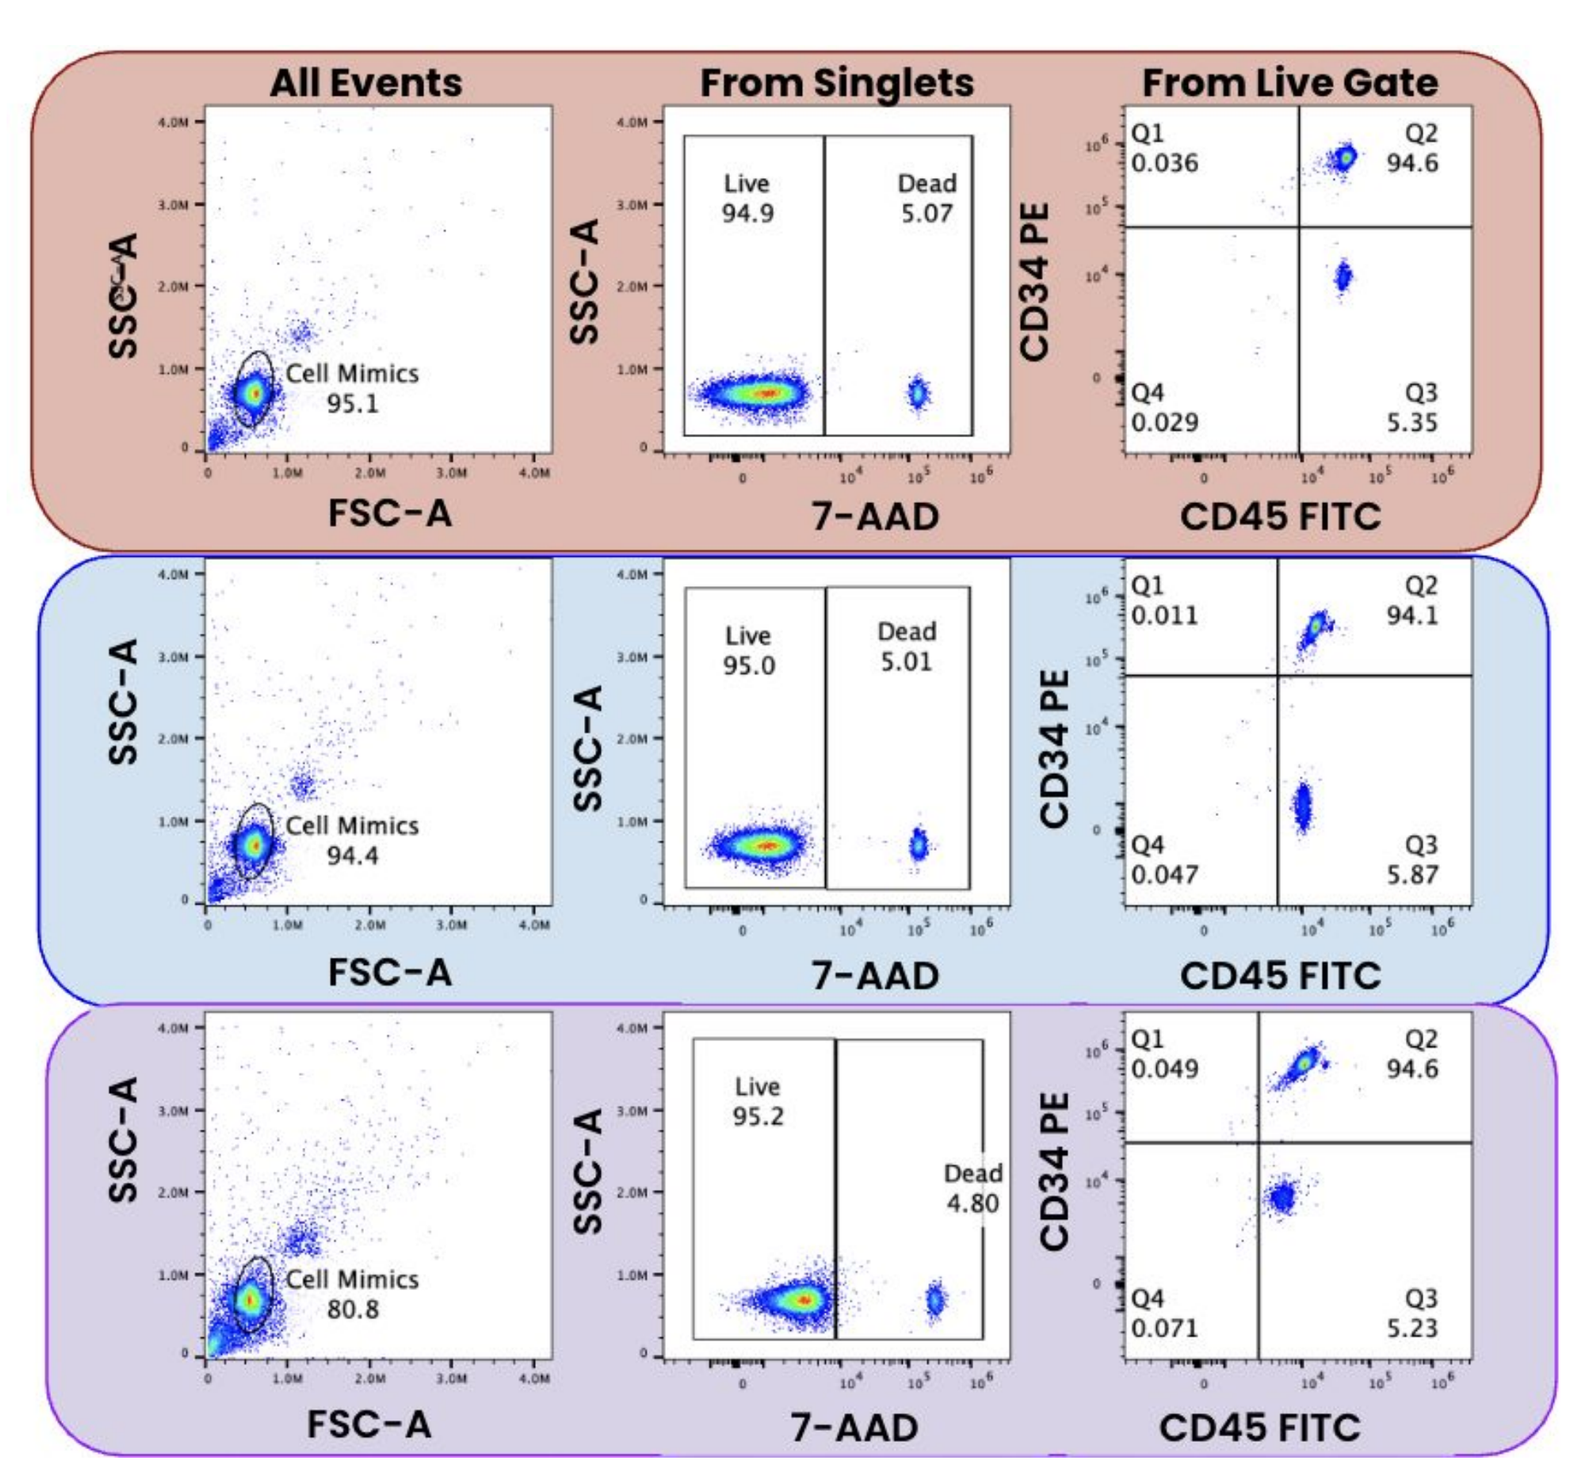

All Events
From Singlets
From Live Gate
Cell Mimics 95.1
Live 94.9
Dead 5.07
Q1 0.036
Q2 94.6
Q4 0.029
Q3 5.35
Cell Mimics 94.4
Live 95.0
Dead 5.01
Q1 0.011
Q2 94.1
Q4 0.047
Q3 5.87
Cell Mimics 80.8
Live 95.2
Dead 4.80
Q1 0.049
Q2 94.6
Q4 0.071
Q3 5.23
FSC-A
7-AAD
CD45 FITC
SSC-A
CD34 PE

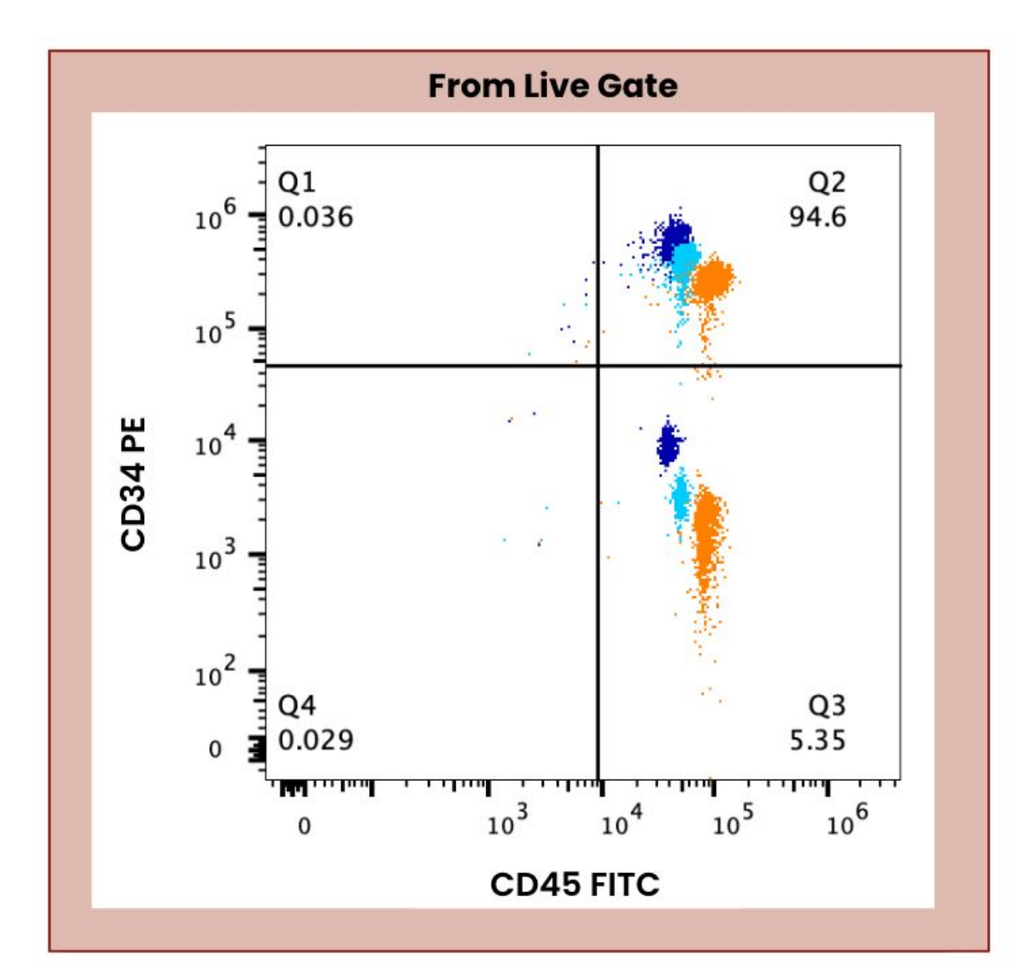

From Live Gate
Q1 0.036
Q2 94.6
Q4 0.029
Q3 5.35
CD34 PE
CD45 FITC

Figure 4. Titrated CD34 and CD45 antibodies from Waters Biosciences. **Dark blue:** 5 µL CD34 + 5 µL CD45; **Light blue:** 2.5 µL CD34 + 5 µL CD45; **Orange:** 1 µL CD34 + 10 µL CD45. Antibody concentrations were titrated to optimize staining performance and reduce background signal and spectral unmixing artifacts.

Figure 3. Comparison of CD34 and CD45 antibodies from three vendors. **Red:** Waters Biosciences; **Blue:** BioLegend; **Purple:** eBiosciences. Vendor-recommended antibody volumes were used; observed background staining and unmixing artifacts may reflect non-optimized antibody titration conditions.

RESULTS: CD34⁺ Stem Cell Mimics Provide Comprehensive Positive and Dead Cell Controls

Population Type	Slingshot Control: CD34+ Stem Cell Mimics	Commercially available controls: Control -1	Commercially available controls: Control -2
CD45+CD34+	Yes	Yes	Yes
Dead (7-AAD)	Yes	Not Recommended	Not Recommended

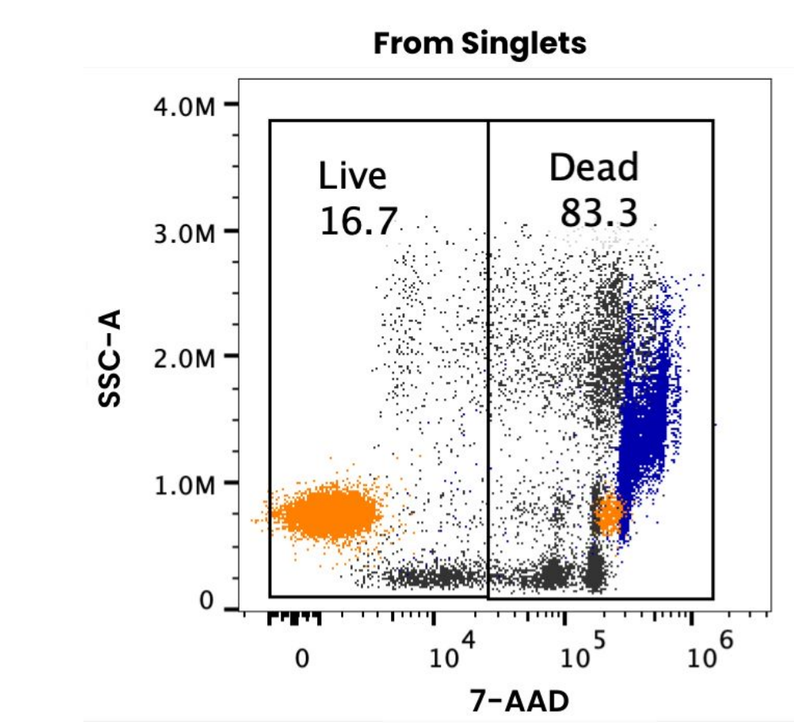

From Singlets
Live 16.7
Dead 83.3
SSC-A
7-AAD

Table 3. (Left) Comparison of control populations available in CD34⁺ Stem Cell Mimics and commercially available controls. **Figure 5.** (Right) Representative overlay of dot plots showing 7-AAD staining in CD34⁺ Stem Cell Mimics (Orange), Control-1 (Blue), and Control-2 (Black).

RESULTS: Spike-in recovery of CD34⁺ Stem Cell Mimics with stabilized whole blood cells and overlay on bone marrow CD34+ Cells

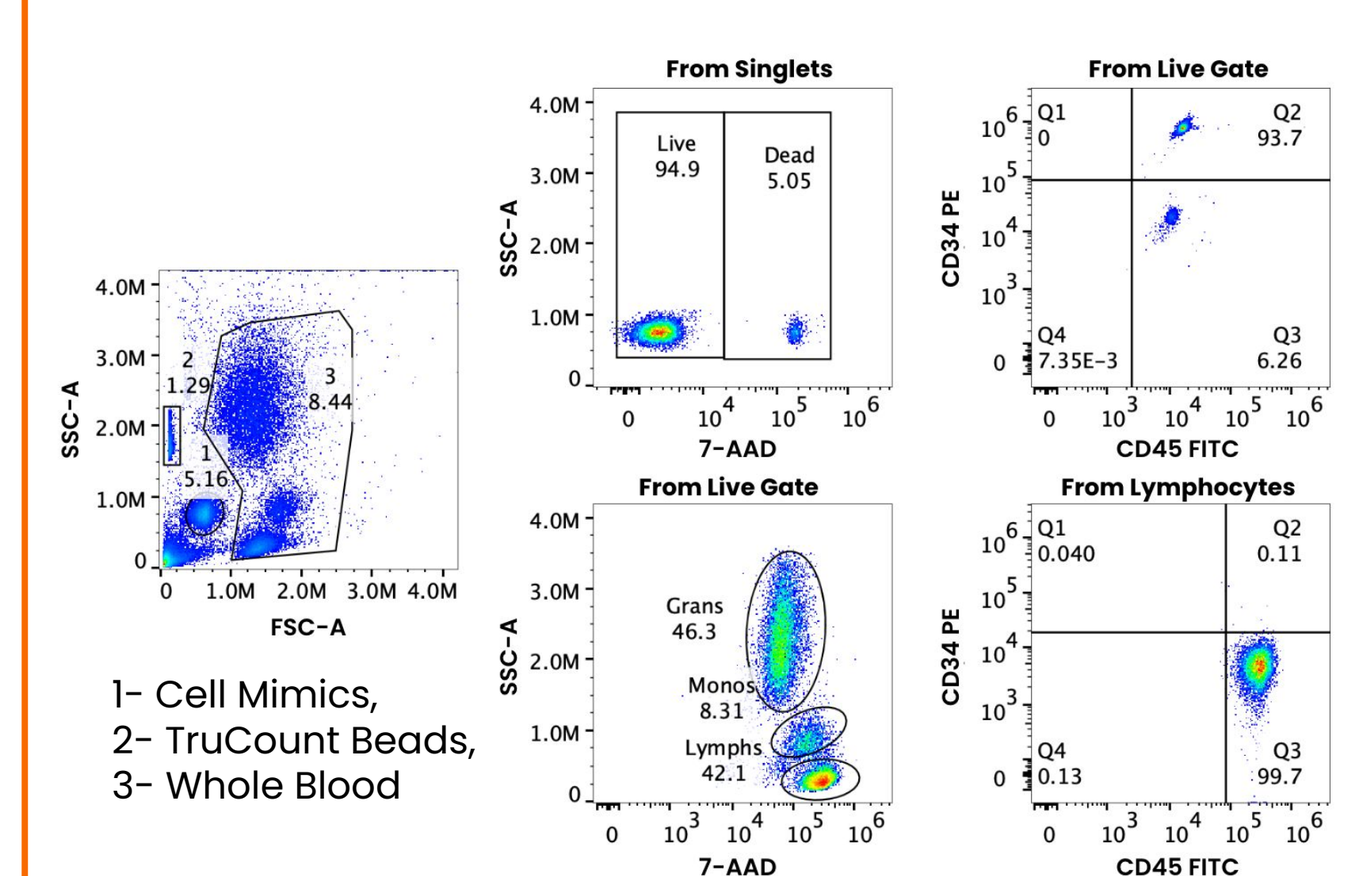

1- Cell Mimics,
2- TruCount Beads,
3- Whole Blood
From Singlets
Live 94.9
Dead 5.05
From Live Gate
Q1 0
Q2 93.7
Q4 7.35E-3
Q3 6.26
Grans 46.3
Monos 8.31
Lymphs 42.1
From Lymphocytes
Q1 0.040
Q2 0.11
Q4 0.13
Q3 99.7
FSC-A
SSC-A
7-AAD
CD34 PE
CD45 FITC

CD45+CD34+ (without 7-AAD)			
Sample	Cell Mimics Count	Bead Count	Mimics /uL
Spike 1	11,461	2,733	1,384
Spike 2	9,370	2,539	1,218
Spike 3	14,458	3,627	1,315
Spike 4	11,756	3,114	1,246

Bead Concentration:49500
Total Volume:150

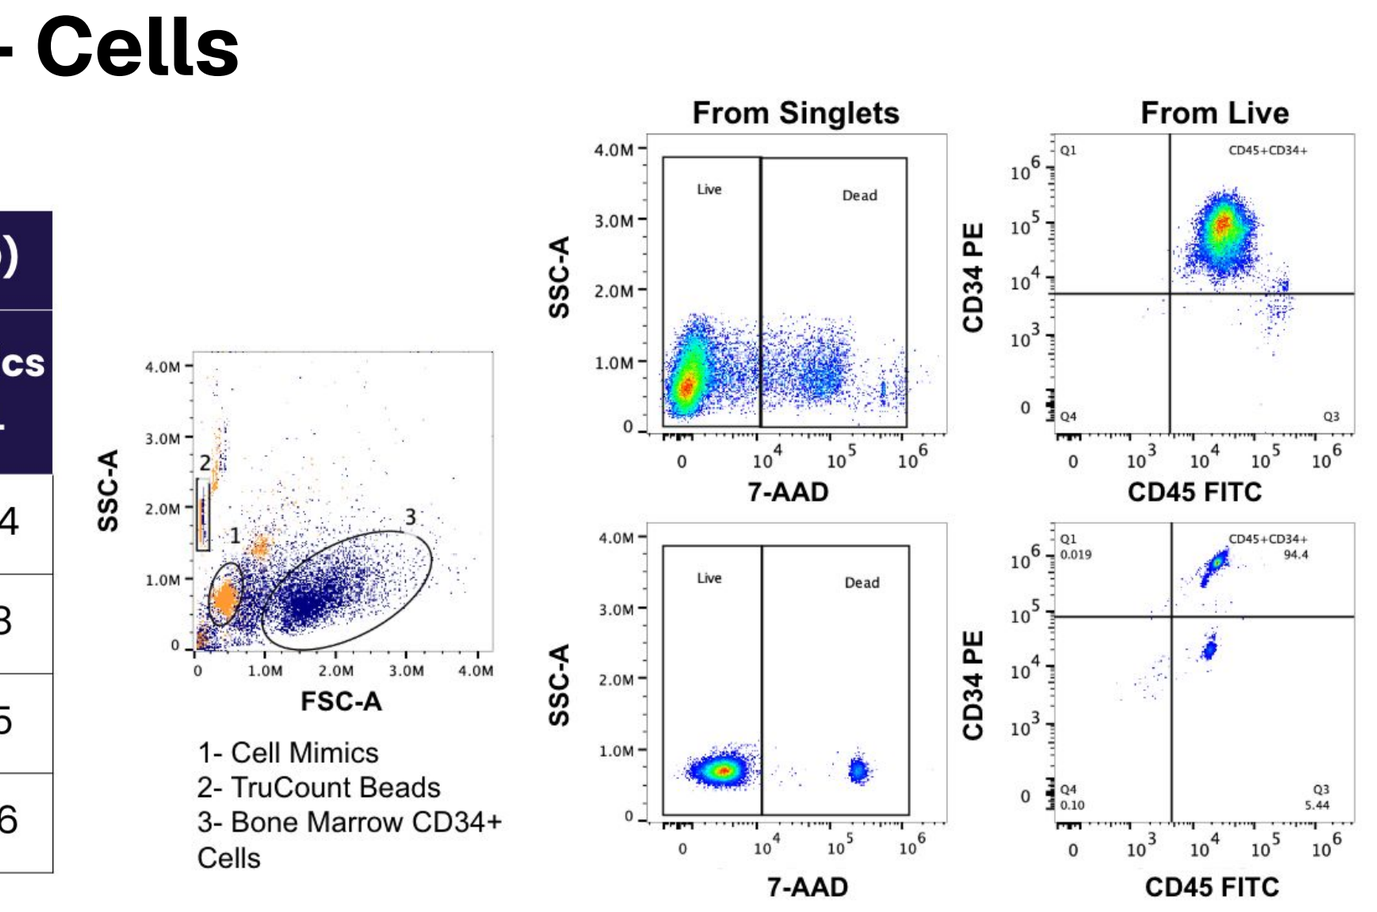

1- Cell Mimics
2- TruCount Beads
3- Bone Marrow CD34+ Cells
From Singlets
From Live
Live
Dead
FSC-A
SSC-A
7-AAD
CD34 PE
CD45 FITC

Figure 6.(Left) representative dot plot schema of CD34⁺ Stem Cell Mimic spike-in analysis using the BD Trucount™ Enumeration Kit. Whole blood was analyzed for CD34⁺ events and Trucount beads for absolute quantification. **Table 4.** (Middle) Summary table of CD45⁺CD34⁺ frequency, recovery, and absolute counts (cells/µL). **Figure 7.**(Right) representative dot plot schema of CD34⁺ Stem Cell Mimic overlay with Bone Marrow CD34+ Cells, analysed using the BD Trucount™ Enumeration Kit.

RESULTS: Controlled Adjustment of CD34⁺ Signal by Dilution with True Negative Cell Mimics

Sample	CD34⁺ Stem Cell Mimics (Total Particles Added)	True Negative Cell Mimics (Total Particles Added)	Dead Cells		CD34+CD45+ Cells		CD45+ Cells	
			Expected	Observed	Expected	Observed	Expected	Observed
No Dilution	125,000	0	5.00	5.01	95.0	94.3	5.00	5.55
Dilution 1	31,250	48,800	1.95	2.16	37.1	37.8	1.95	2.39
Dilution 2	1,953	48,800	0.19	0.27	3.66	3.94	0.19	0.55

Table 5. Controlled modulation of CD34⁺ Stem Cell Mimic and cell population signals by dilution with True Negative Cell Mimics. Expected and observed values are shown across three dilution conditions.

SUMMARY

- CD34⁺ Stem Cell Mimics are compatible with the ISHAGE gating strategy.
- CD34⁺ Stem Cell Mimics show high precision with low variability across runs, operators, and instruments.
- CD34⁺ Stem Cell Mimics showed high closed-vial stability over 24 months when assessed under accelerated conditions at 37 °C using the Arrhenius model.
- Spike-in studies confirm that CD34⁺ Stem Cell Mimics can be directly spiked into clinical samples for routine flow cytometry analysis.
- Dilution studies enable customizable CD34⁺ levels using True Negative Cell Mimics to meet laboratory-specific requirements.

SlingshotBio™

ISO 9001:2015 Certified
ISO 17034 & ISO 13485 standards aligned